Effects of the oral, small molecule GLP-1R Agonist, TTP273, on patients with Stage 2 hypertension: Results from a post-hoc analysis of the phase 2 LOGRA study

Carmen Valcarce, Imogene Dunn and Jennifer Freeman, vTv Therapeutics LLC, High Point, NC, USA

Introduction

TTP273, an oral, non-peptide GLP-1R agonist showed significant placebo reductions in A1c (0.9 and 0.7% when dosed at 150mg QPM or BID, respectively) in the LOGRA study, a 12-week, phase 2, double-blind, placebo-controlled randomized trial. In addition, less nausea was observed in the active groups than in the placebo group, and the only incident of vomiting occurred in the placebo group.

Post-hoc analysis of the data reveal further information about the effects of TTP273.
Herein we present the results of a post-hoc analysis of patients from the LOGRA study with Stage 2 hypertension.

Aim

The goal of this post-hoc analysis was to examine the effects following completion of 12 weeks of treatment with TTP273 in patients with Stage 2 hypertension at study start.

LOGRA Study Design

- 174 T2DM patients randomized
- 7.5-10% baseline HbA1c
- BMI 25-45 kg/m^2
- Arms (1:1:1)
 - Placebo (PBO)
 - TTP273 150mg once daily (QPM)
 - TTP273 150mg twice daily (BID)

- Entry criteria allowed patients on standard of care treatment for hypertension, but excluded patients with persistent, uncontrolled hypertension at screening (SBP ≥ 160mmHg and/or diastolic blood pressure (DBP) ≥ 90mmHg).

Demography – Baseline Characteristics

Approximately 25% of the patients that completed the LOGRA study were considered to have Stage 2 hypertension at baseline (defined by SBP ≥ 140mmHg or DBP ≥90mmHg).

Baseline characteristics among treatment groups were relatively well balanced with similar baseline values (mean baseline values of 146/85 mmHg and 72 bpm, for SBP/DBP and pulse rate respectively).

At baseline, approximately 70% of the patients with Stage 2 hypertension received 1 or more concomitant medications to treat hypertension. No changes in these medications were made during the study, with the exception of one placebo patient.

	PBO n=9	150mg BID n=11	150 mg QPM n=9
Age (years): Mean (median)	60 (58)	56 (57)	58 (61)
Gender: M to F	5:4	6:5	4:5
Ethnicity: Hisp: Not Hispanic	2:7	6:5	2:7
Race: White/AA/Asian	7/2/0	7/4/0	5/3/1
Weight (kg) mean (median)	101 (100)	102 (95)	89 (84)
Baseline HbA1c (%) mean (median)	8.4 (8.3)	8.1 (8.0)	8.5 (8.6)
Baseline SBP (mmHg) mean (median)	143 (147)	148 (146)	148 (147)
Baseline DBP (mmHg) mean (median)	83 (83)	86 (88)	86 (86)
Baseline Pulse rate (beats/min)	76 (76)	69 (68)	72 (78)
1 or more con meds to treat hypertension (patients, n (%))	6 (67%)	7 (64%)	7 (78%)

Results: Subgroup Analysis LOGRA STUDY by ACC/AHA Blood Pressure Category at Baseline

Patients were grouped based on their baseline blood pressure values according to the 2017 ACC/AHA High Blood Pressure Guidelines

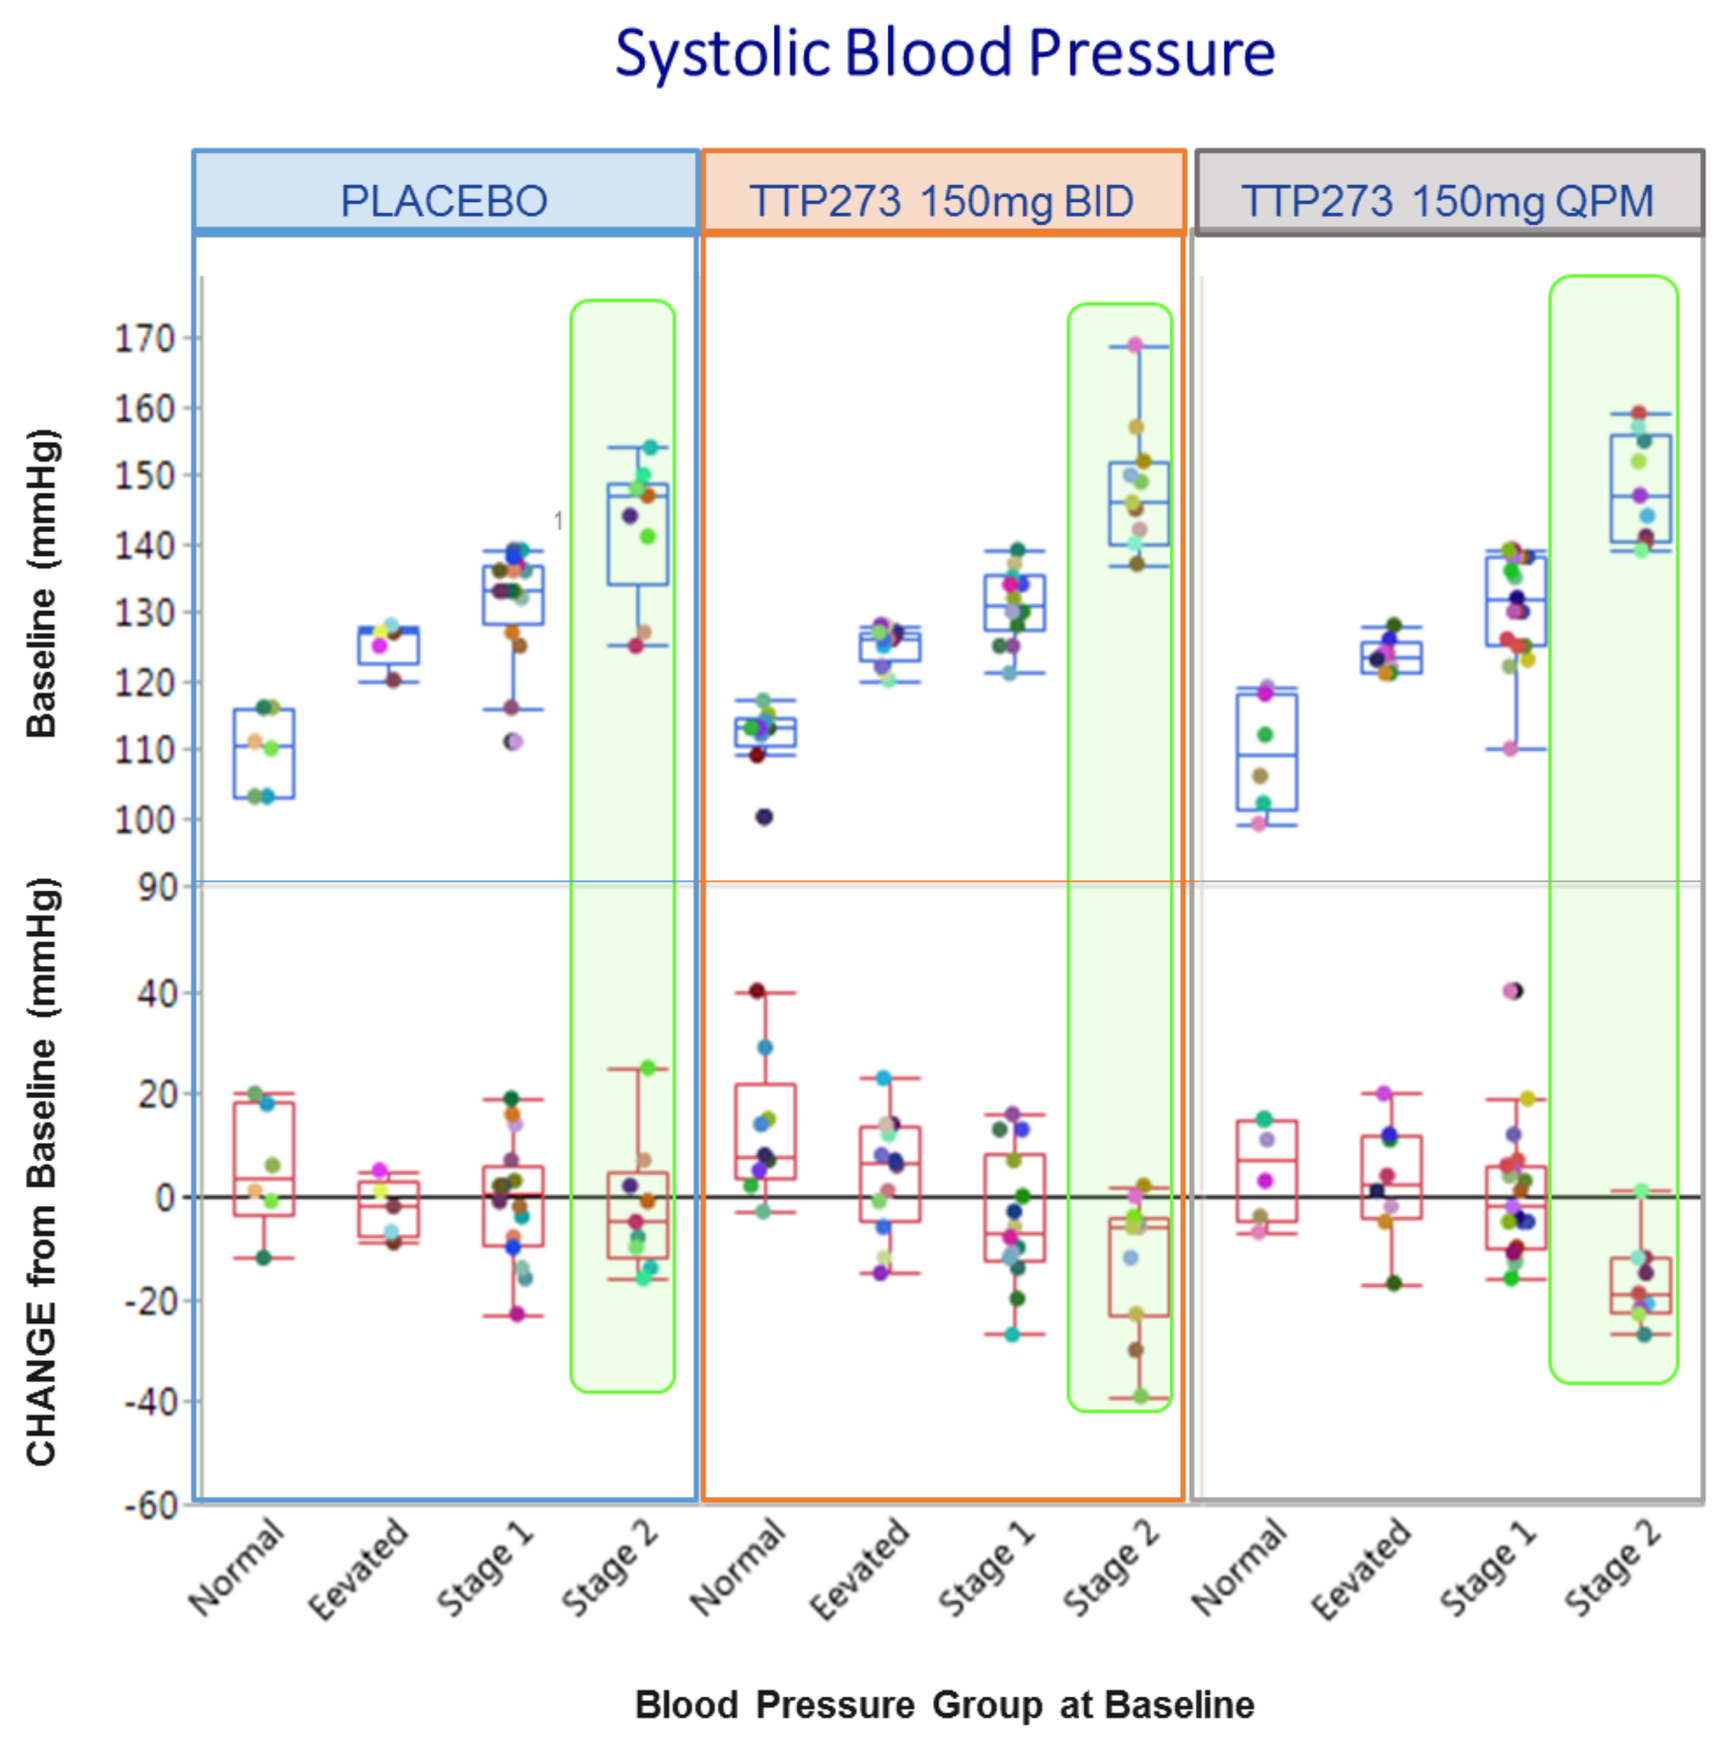
Systolic Blood Pressure
PLACEBO
TTP273 150mg BID
TTP273 150mg QPM
Baseline (mmHg)
CHANGE from Baseline (mmHg)
Normal
Eevated
Stage 1
Stage 2
Blood Pressure Group at Baseline

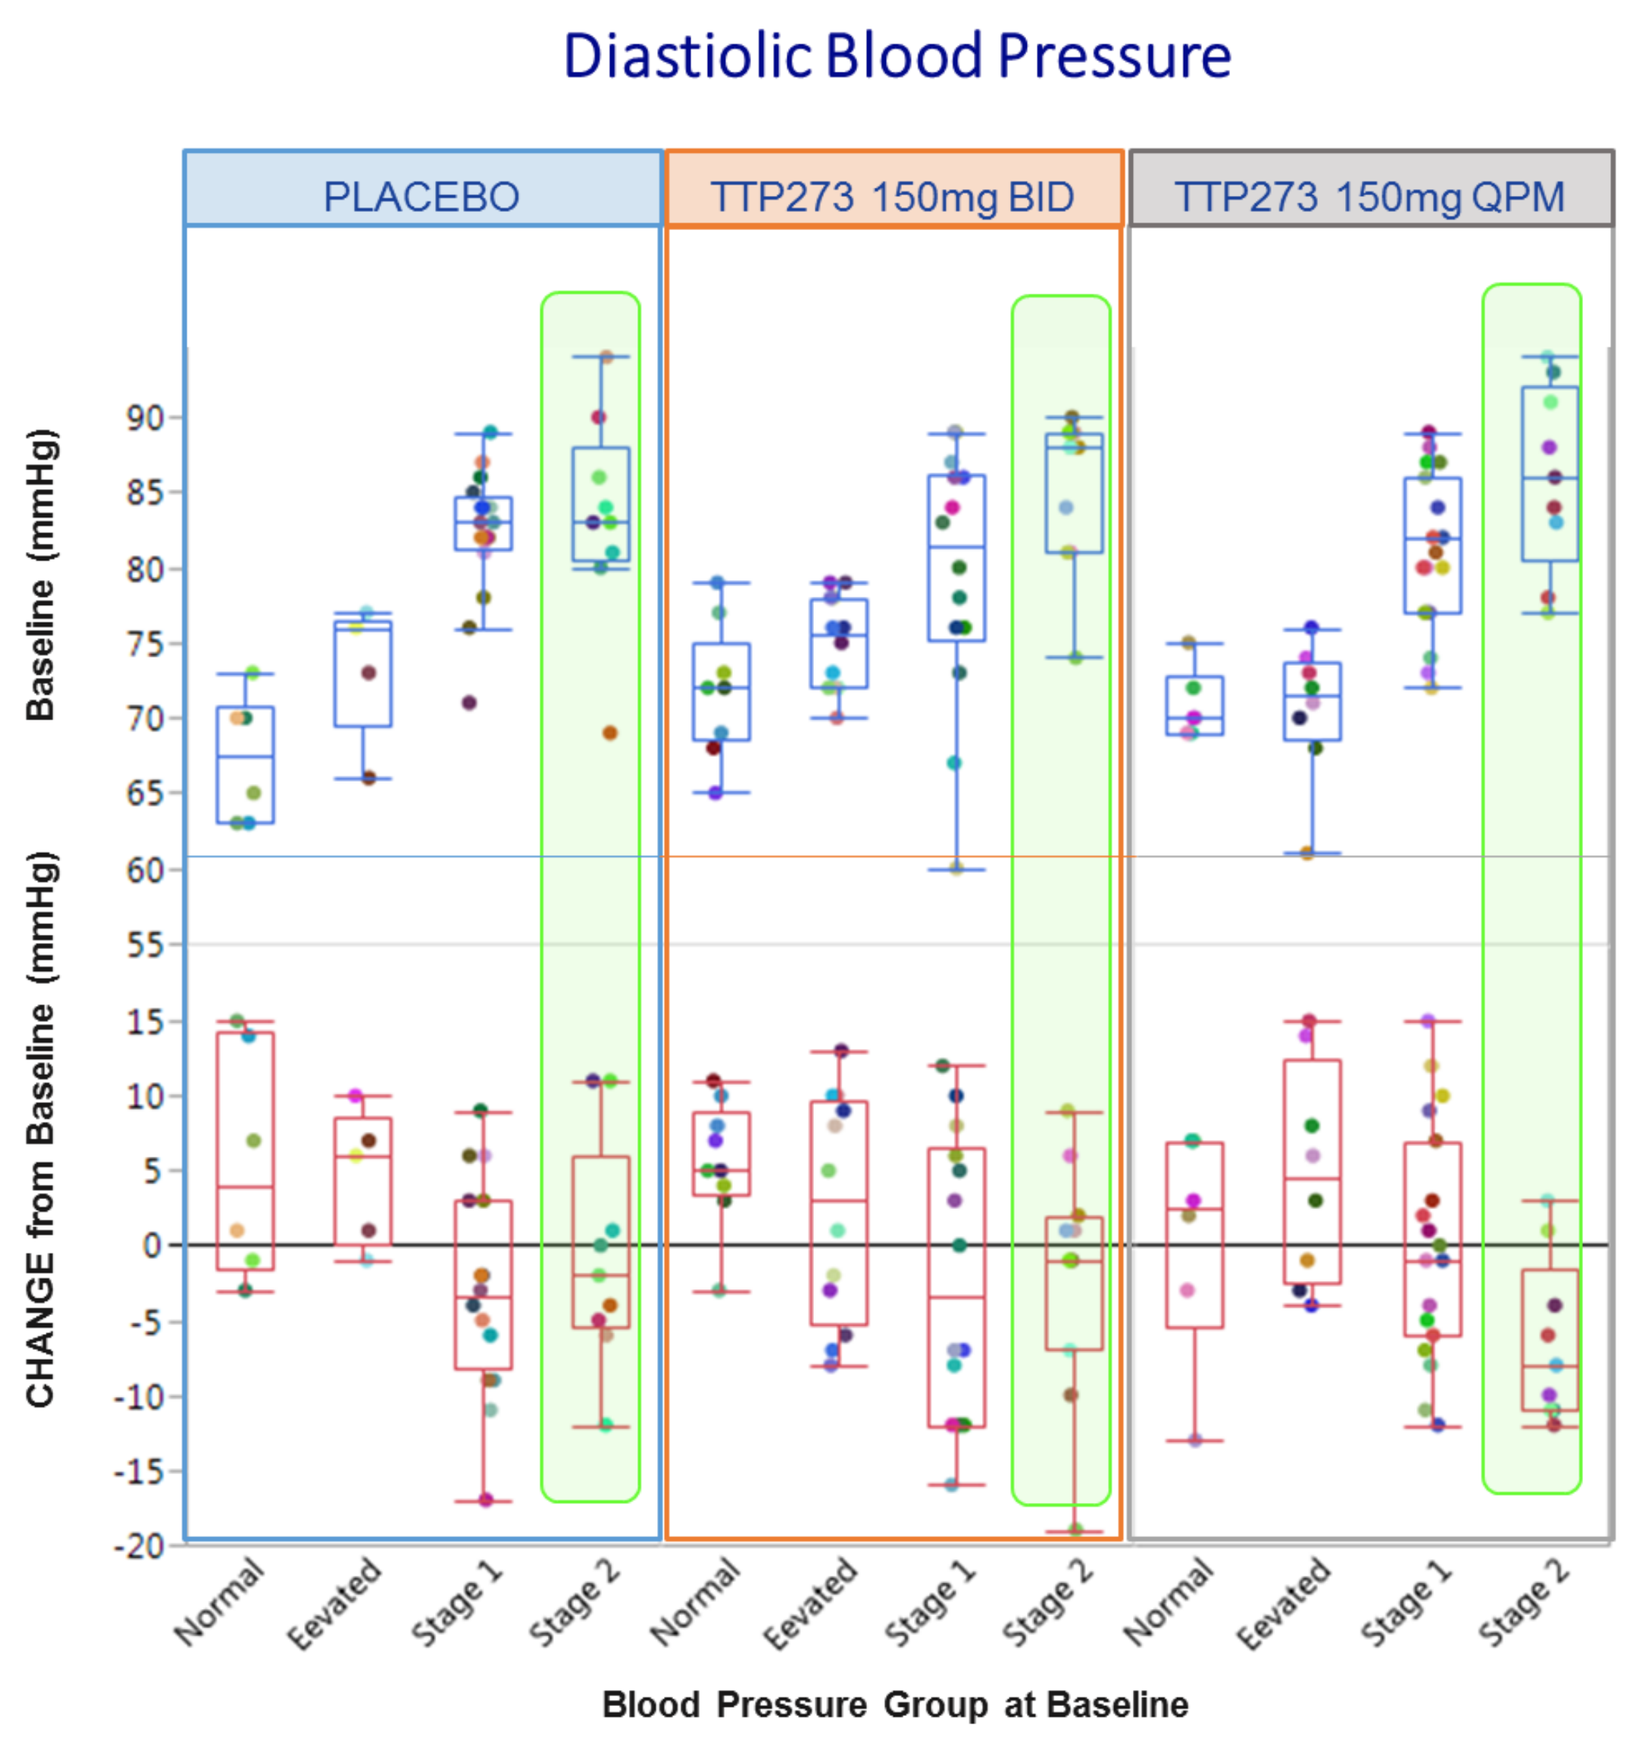
Diastiolic Blood Pressure
PLACEBO
TTP273 150mg BID
TTP273 150mg QPM
Baseline (mmHg)
CHANGE from Baseline (mmHg)
Normal
Eevated
Stage 1
Stage 2
Blood Pressure Group at Baseline

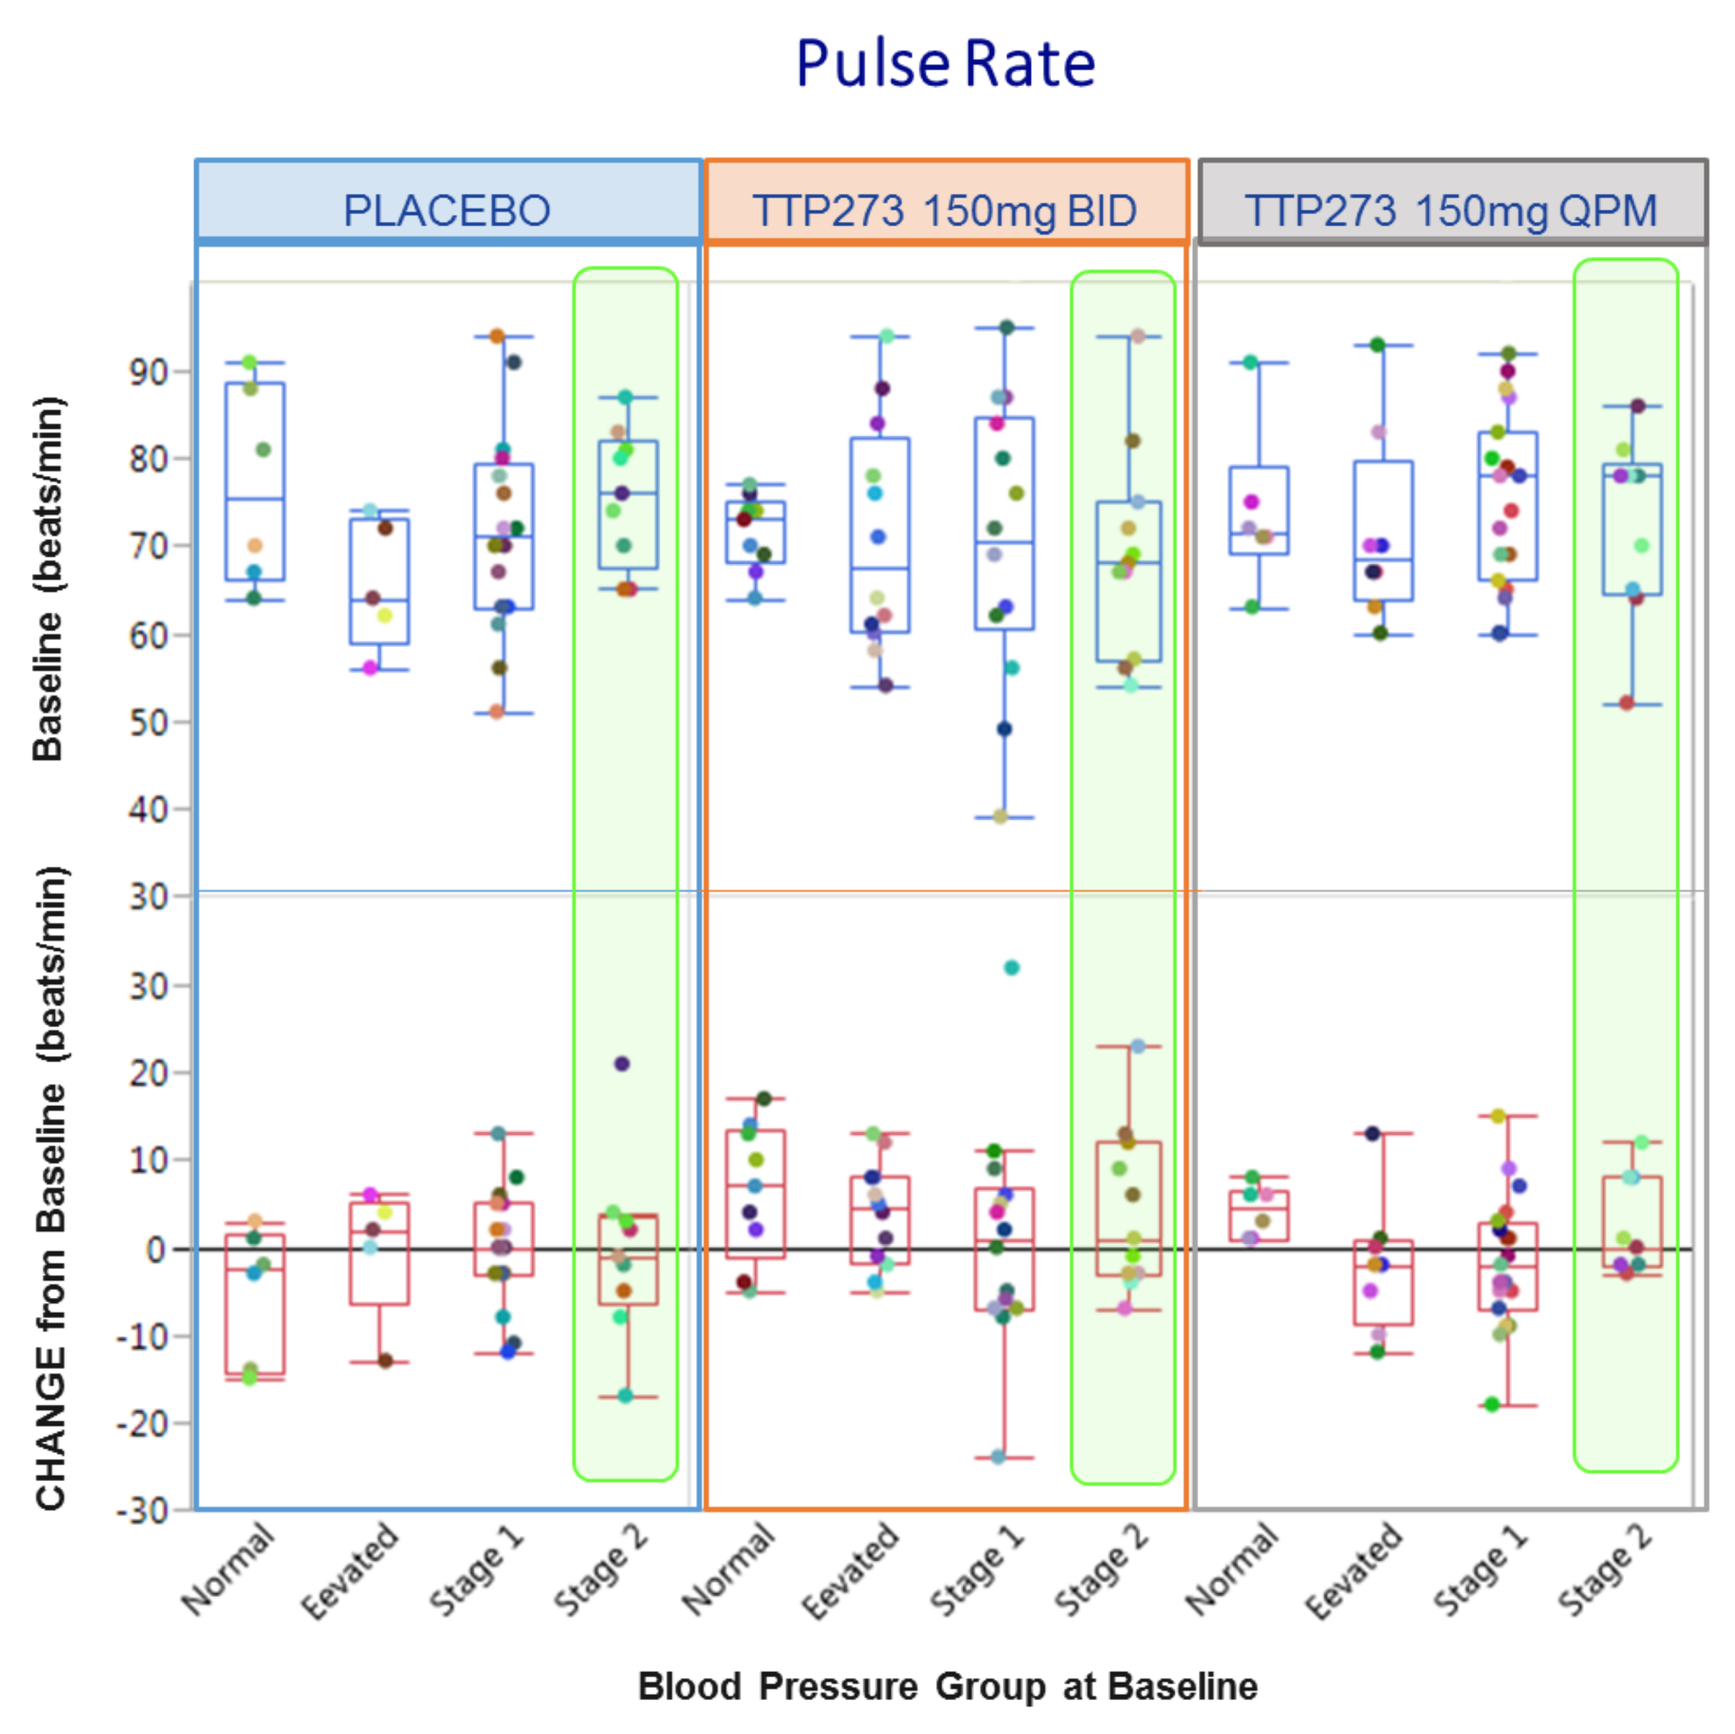
Pulse Rate
PLACEBO
TTP273 150mg BID
TTP273 150mg QPM
Baseline (beats/min)
CHANGE from Baseline (beats/min)
Normal
Eevated
Stage 1
Stage 2
Blood Pressure Group at Baseline

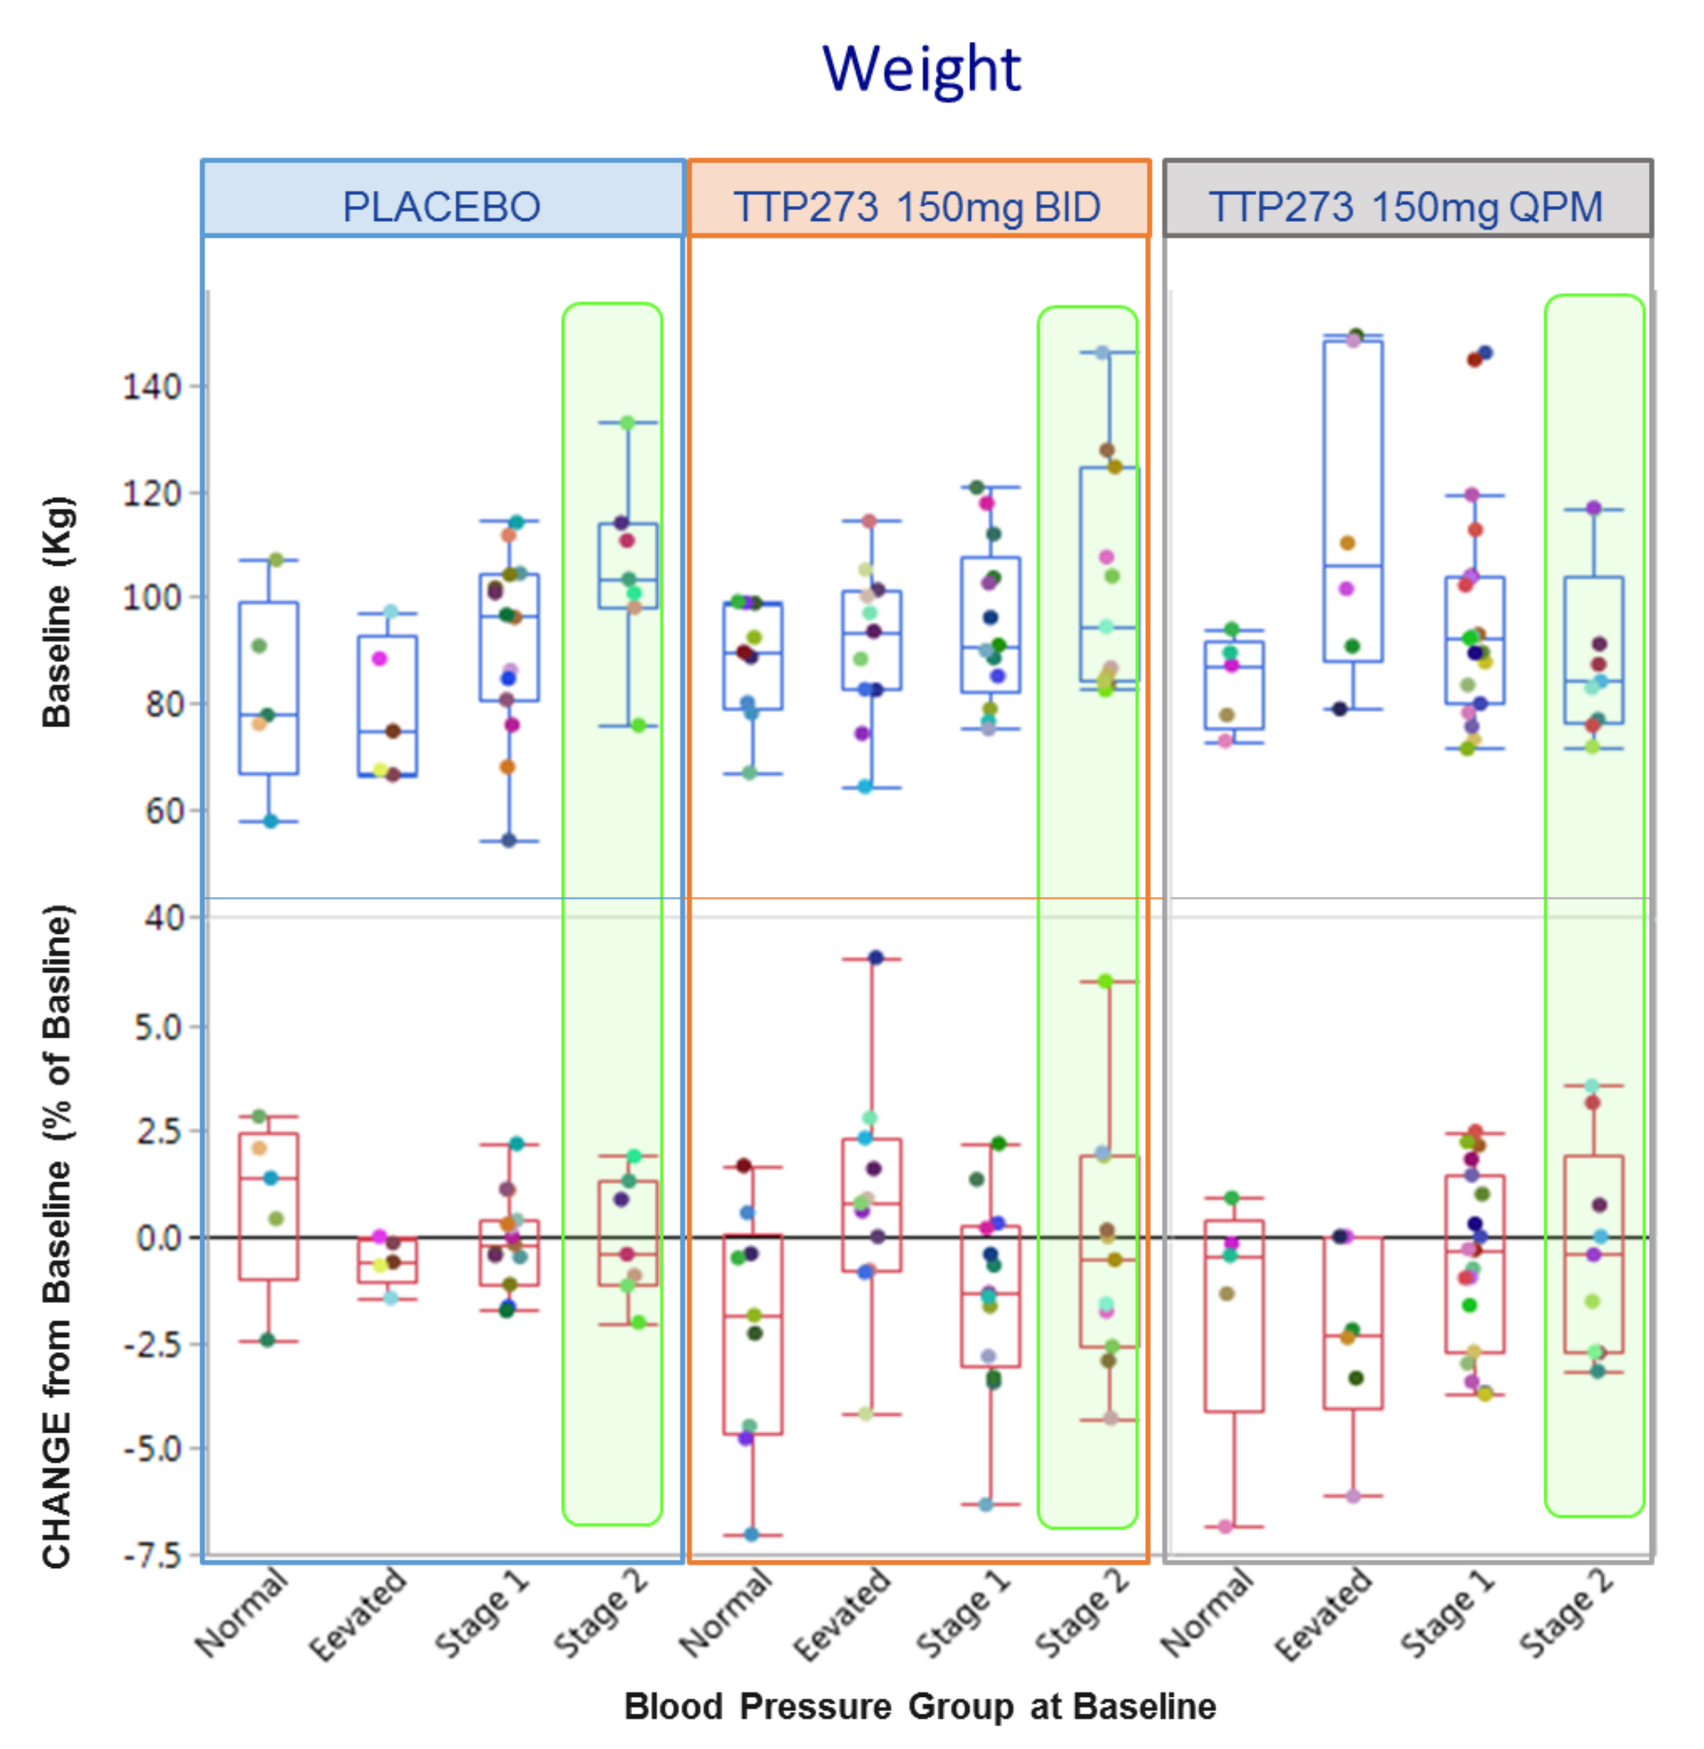
Weight
PLACEBO
TTP273 150mg BID
TTP273 150mg QPM
Baseline (Kg)
CHANGE from Baseline (% of Basline)
Normal
Eevated
Stage 1
Stage 2
Blood Pressure Group at Baseline

- Within each blood pressure category, baseline values were reasonably comparable among treatment groups.
- Post-hoc analysis of subjects with Stage 2 hypertension showed reductions in SBP and DBP for subjects treated with TTP273, with more pronounced decreases observed with QPM versus BID dosing.
- A nominally statistically significant decrease from baseline in SBP of 17 mmHg (p = 0.01 versus placebo) was observed for subjects treated with TTP273 150 mg QPM.
- Lack of associated changes in bodyweight indicate blood pressure response is not driven by changes in bodyweight.
- No consistent trends related to blood pressure categories were observed for pulse.

Blood Pressure Categories

BLOOD PRESSURE CATEGORY	SYSTOLIC mm Hg (upper number)		DIASTOLIC mm Hg (lower number)
NORMAL	LESS THAN 120	and	LESS THAN 80
ELEVATED	120 – 129	and	LESS THAN 80
HIGH BLOOD PRESSURE (HYPERTENSION) STAGE 1	130 – 139	or	80 – 89
HIGH BLOOD PRESSURE (HYPERTENSION) STAGE 2	140 OR HIGHER	or	90 OR HIGHER

American Heart Association | American Stroke Association

Change from Baseline Vital Signs for Patients with Stage 2 hypertension

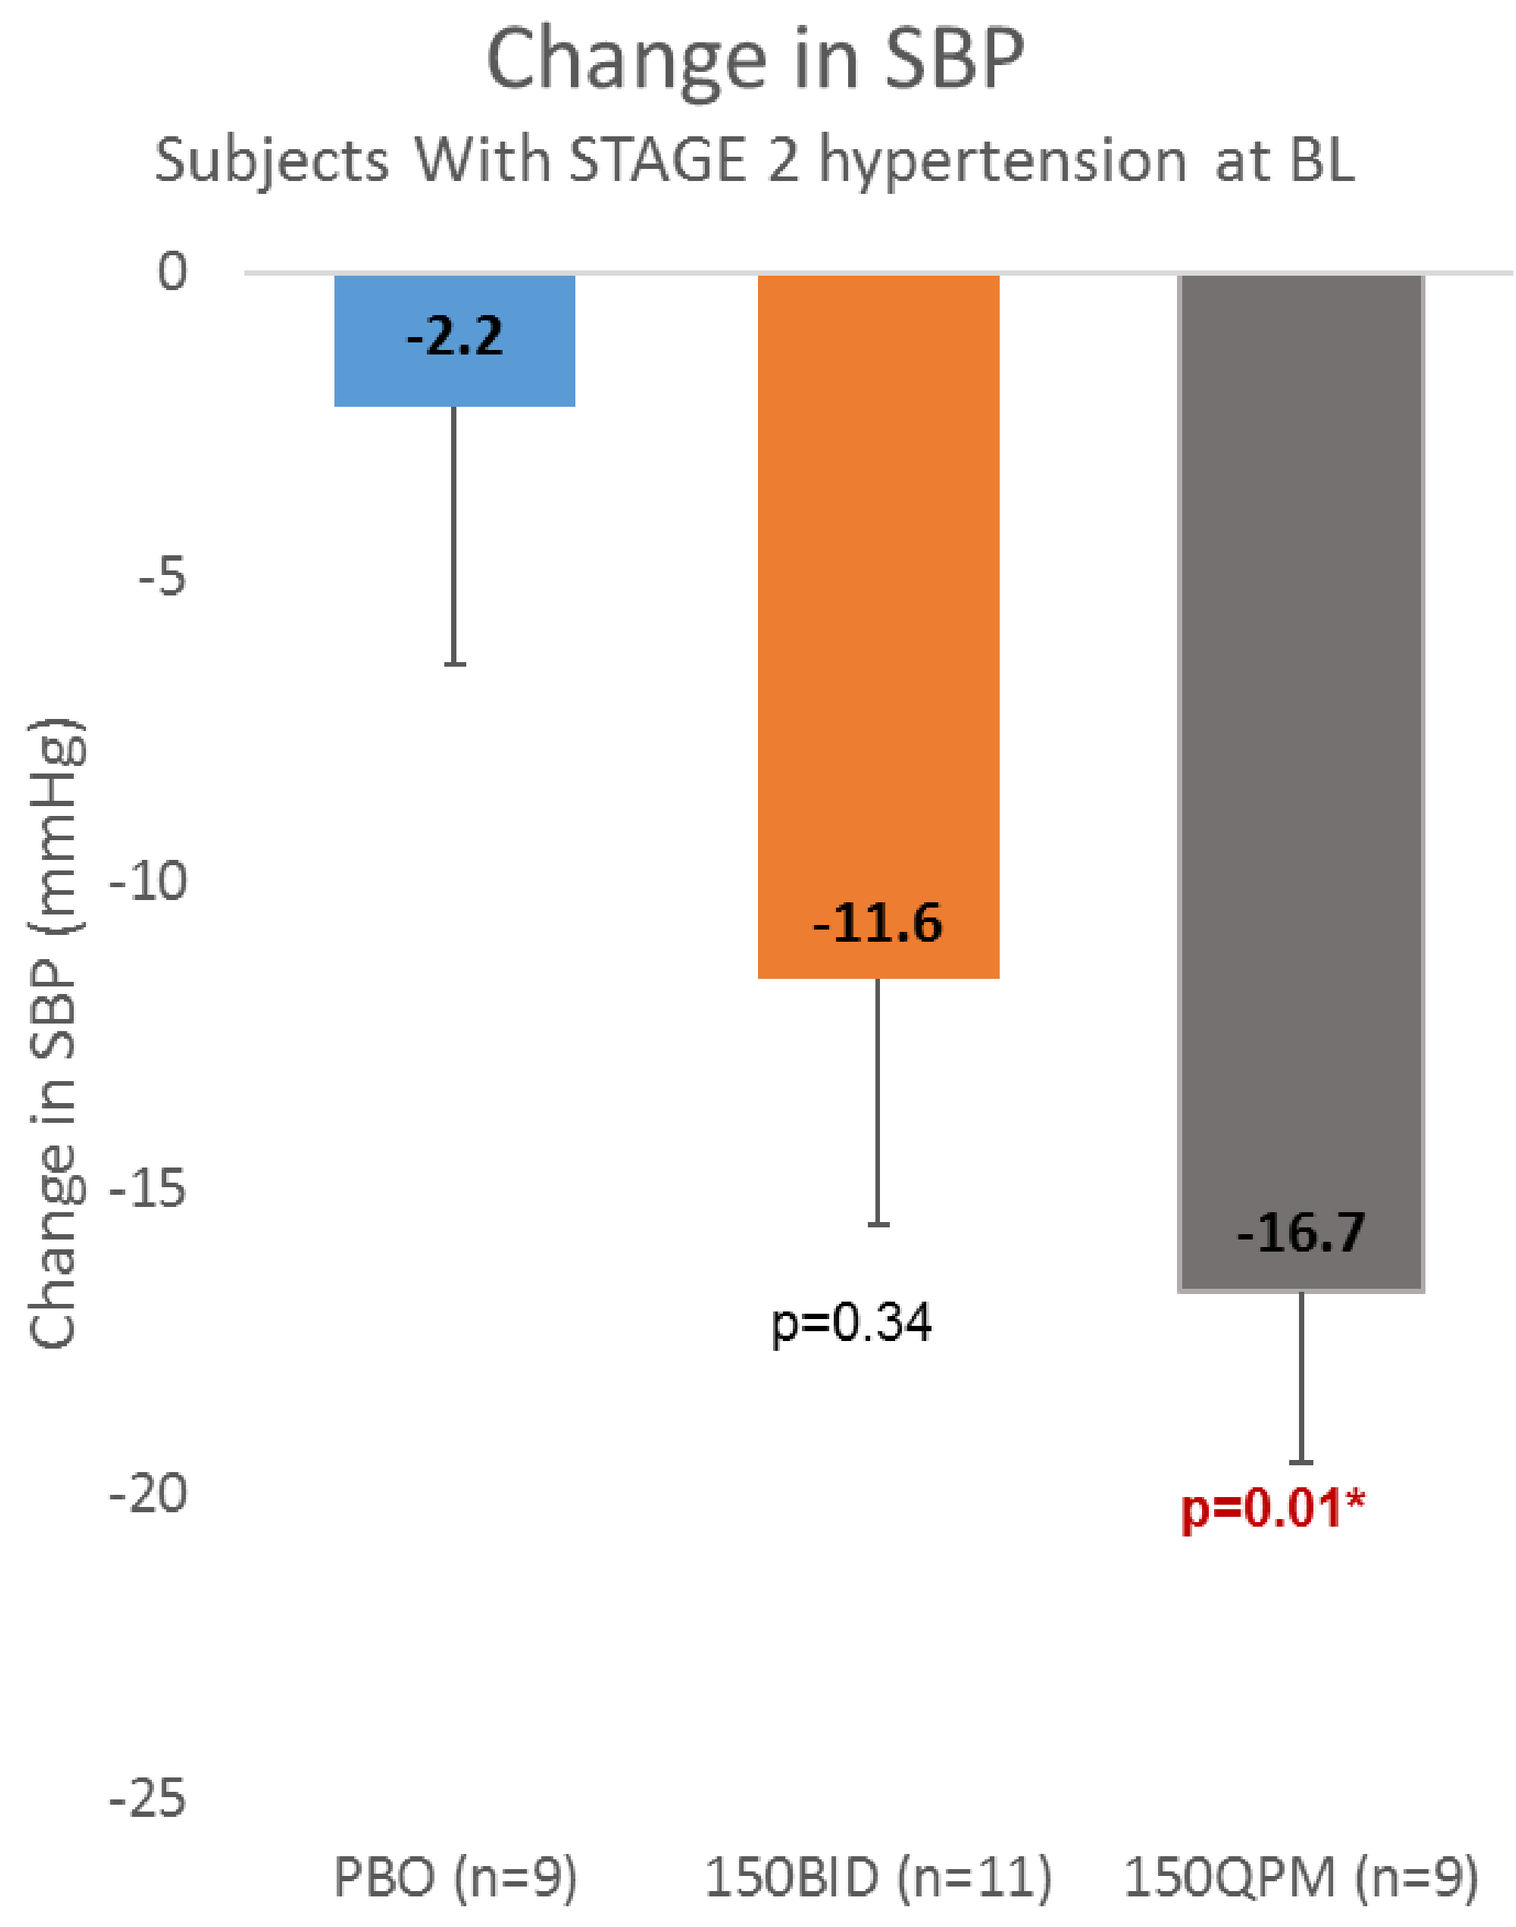
Change in SBP
Subjects With STAGE 2 hypertension at BL
Change in SBP (mmHg)
-2.2
-11.6
-16.7
p=0.34
p=0.01*
PBO (n=9)
150BID (n=11)
150QPM (n=9)

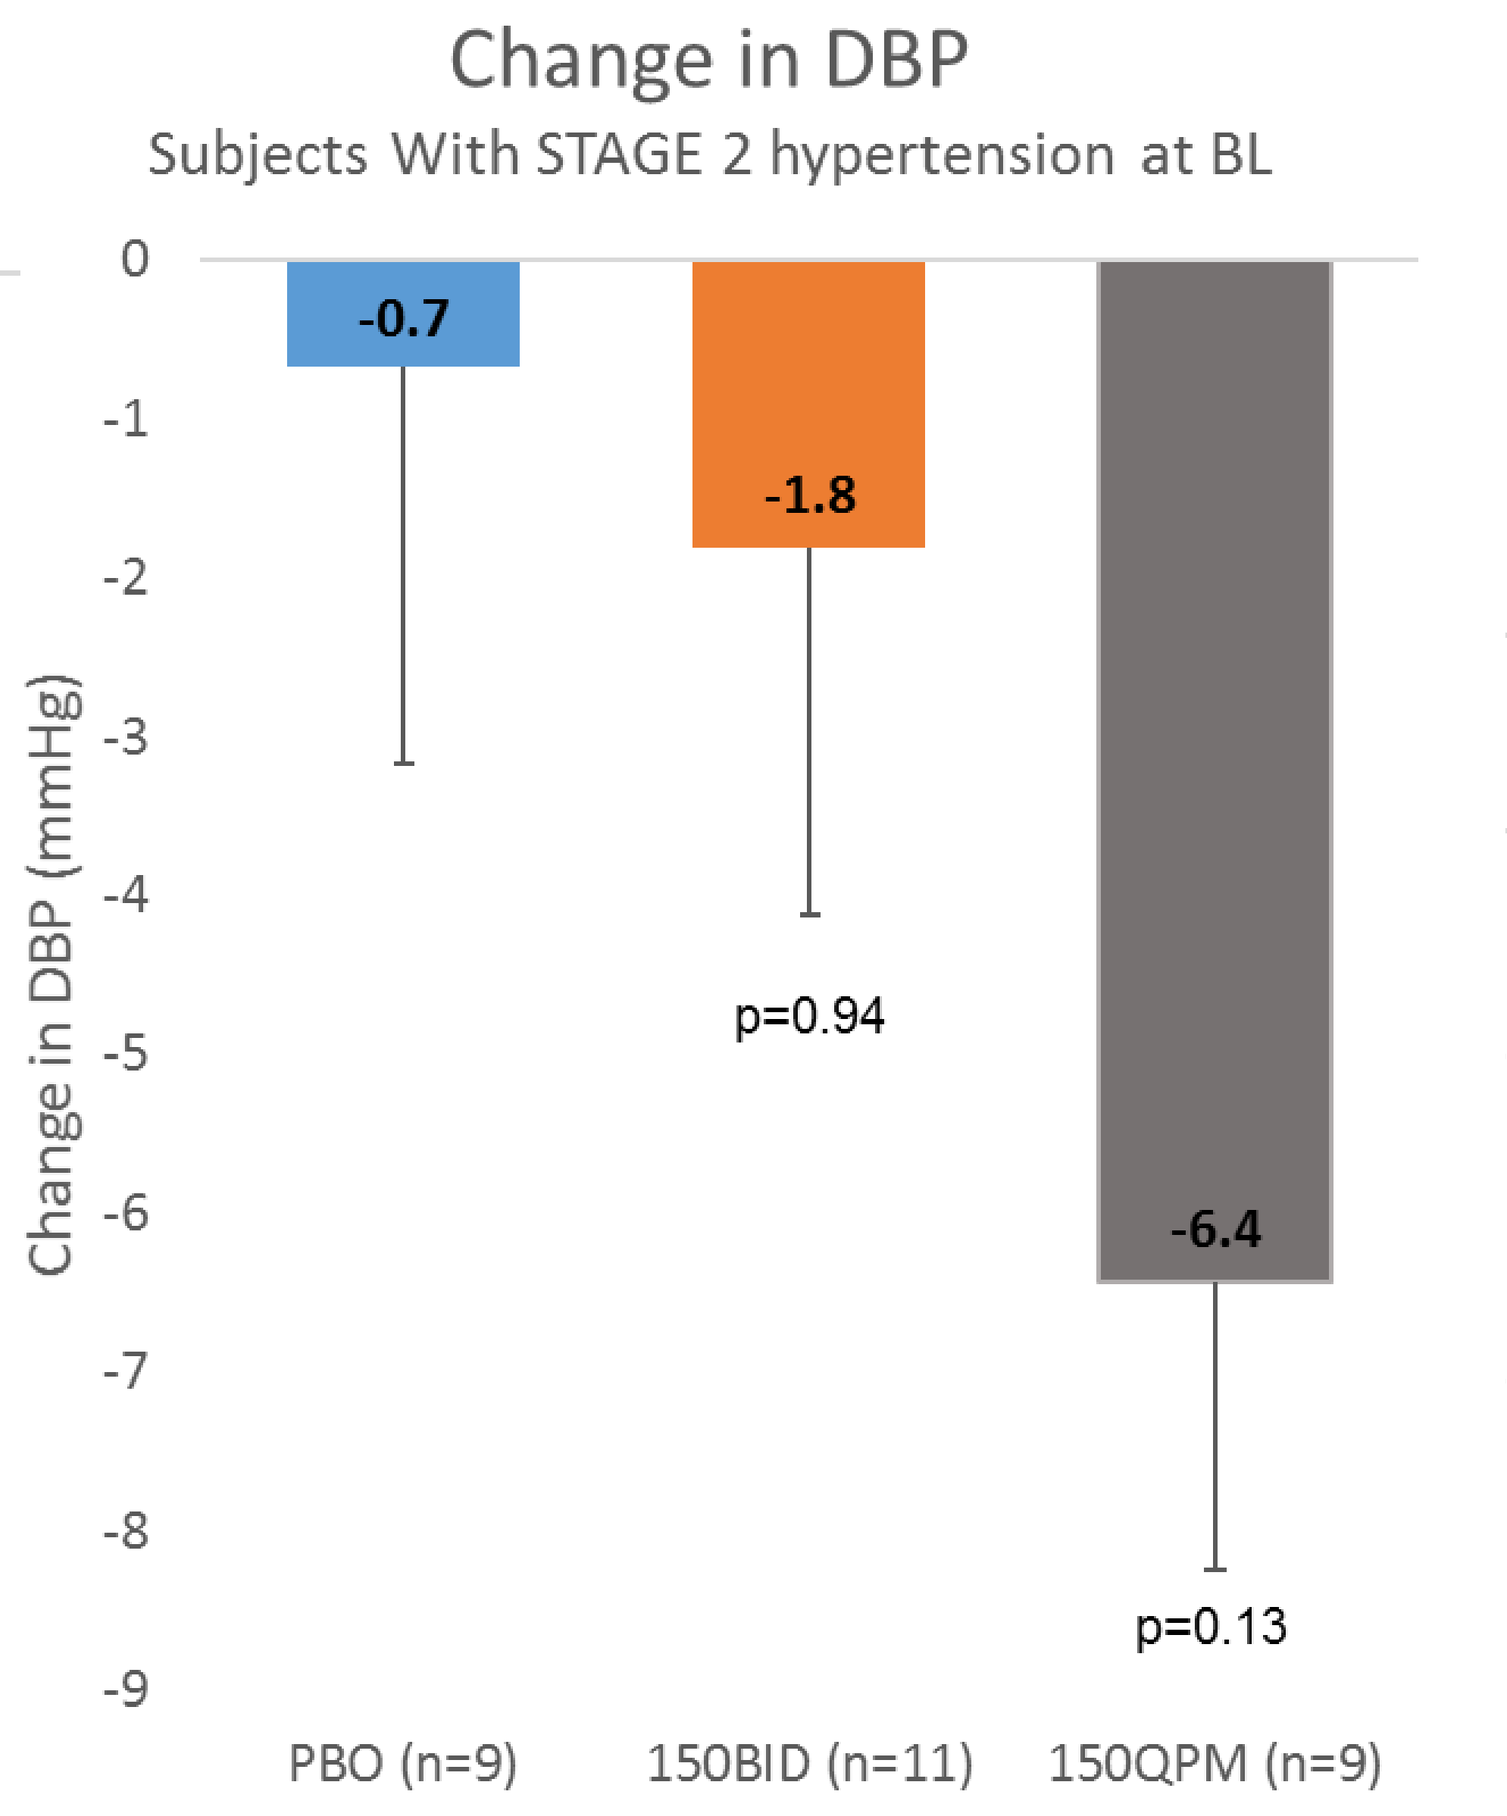
Change in DBP
Subjects With STAGE 2 hypertension at BL
Change in DBP (mmHg)
-0.7
-1.8
-6.4
p=0.94
p=0.13
PBO (n=9)
150BID (n=11)
150QPM (n=9)

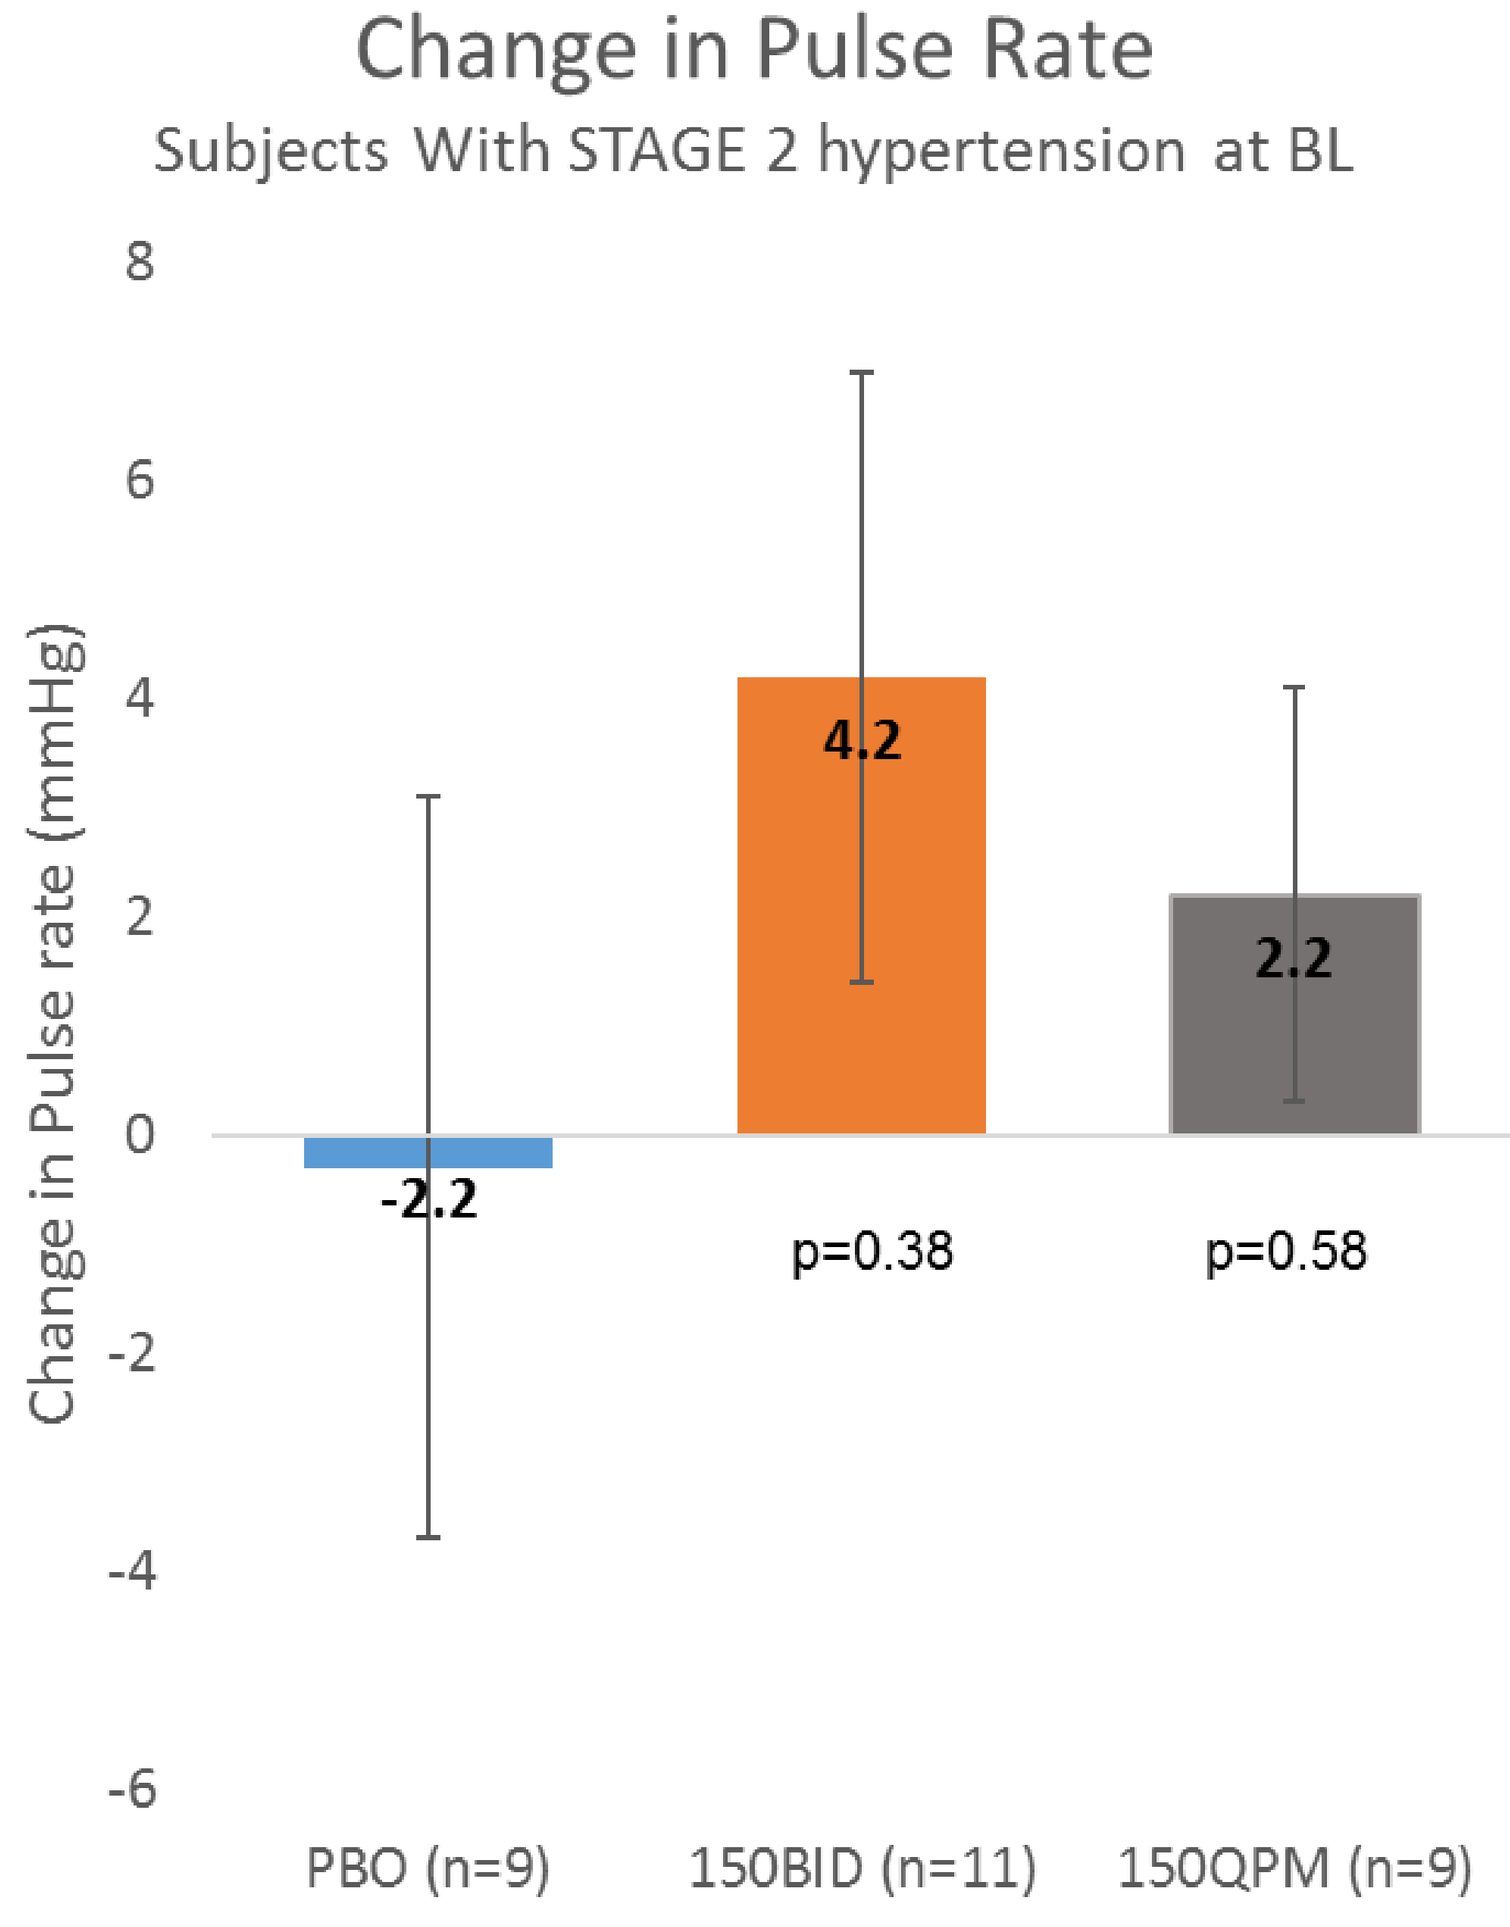
Change in Pulse Rate
Subjects With STAGE 2 hypertension at BL
Change in Pulse rate (mmHg)
-2.2
4.2
2.2
p=0.38
p=0.58
PBO (n=9)
150BID (n=11)
150QPM (n=9)

CONCLUSIONS

- This Phase 2a study confirmed the potential of TTP273 as a treatment for Type 2 diabetes that could potentially expand the use of the GLP-1 therapeutic class.
- Results from this post-hoc analysis suggest that TTP273 could provide an additional benefit of reducing SBP, in line with other GLP-1R agonists.
- Unlike other GLP-1R agonists, these effects occurred without the side effects of nausea and vomiting.
- Further studies are needed to determine the optimal dose/dose regimen and/or target population