Effect of CYP2C8 and CYP3A4 inhibition and CYP induction on the pharmacokinetics of azeliragon

Aaron H Burstein[1], Michael J Lamson[2], Mark Sale[2], Scott J Brantley[2], Ann Gooch[1], Imogene Dunn[1], Larry Altstiel[1]
(1) vTv Therapeutics LLC, High Point, NC; (2) Nuventra Pharma Sciences, Inc. Durham, NC

Introduction

Azeliragon is an oral antagonist of the Receptor for Advanced Glycation Endproducts (RAGE) currently being evaluated in a pivotal Phase 3 study for mild Alzheimer's disease (AD). Enzyme phenotyping suggested CYP2C8 and CYP3A4 were the predominant enzymes responsible for the metabolism of azeliragon. The present study evaluated the effect of CYP2C8 and CYP3A4 inhibition and CYP induction on the pharmacokinetics of azeliragon and its major metabolites (M1, M2, M3).

Objectives

The primary objectives of this study were to :
- Evaluate the effect of gemfibrozil (CYP2C8 inhibition) on the PK of azeliragon.
- Evaluate the effect of nefazodone (CYP3A4 inhibition) on the PK of azeliragon.
- Evaluate the effect of rifampin (CYP induction) on the PK of azeliragon.

The secondary objectives of this study were to:
- Evaluate the effects of gemfibrozil, nefazodone, and rifampin on the PK of azeliragon metabolites of interest (M1, M2 and M3).

Study Design

- Randomized, open-label, multiple-dose, parallel design trial in which a total of 75 subjects received azeliragon orally either alone (20 subjects) or in combination with gemfibrozil (18 subjects), nefazodone (18 subjects), or rifampin (19 subjects).
- For the purposes of the PK analysis, the cohorts were characterized into 4 treatment regimens

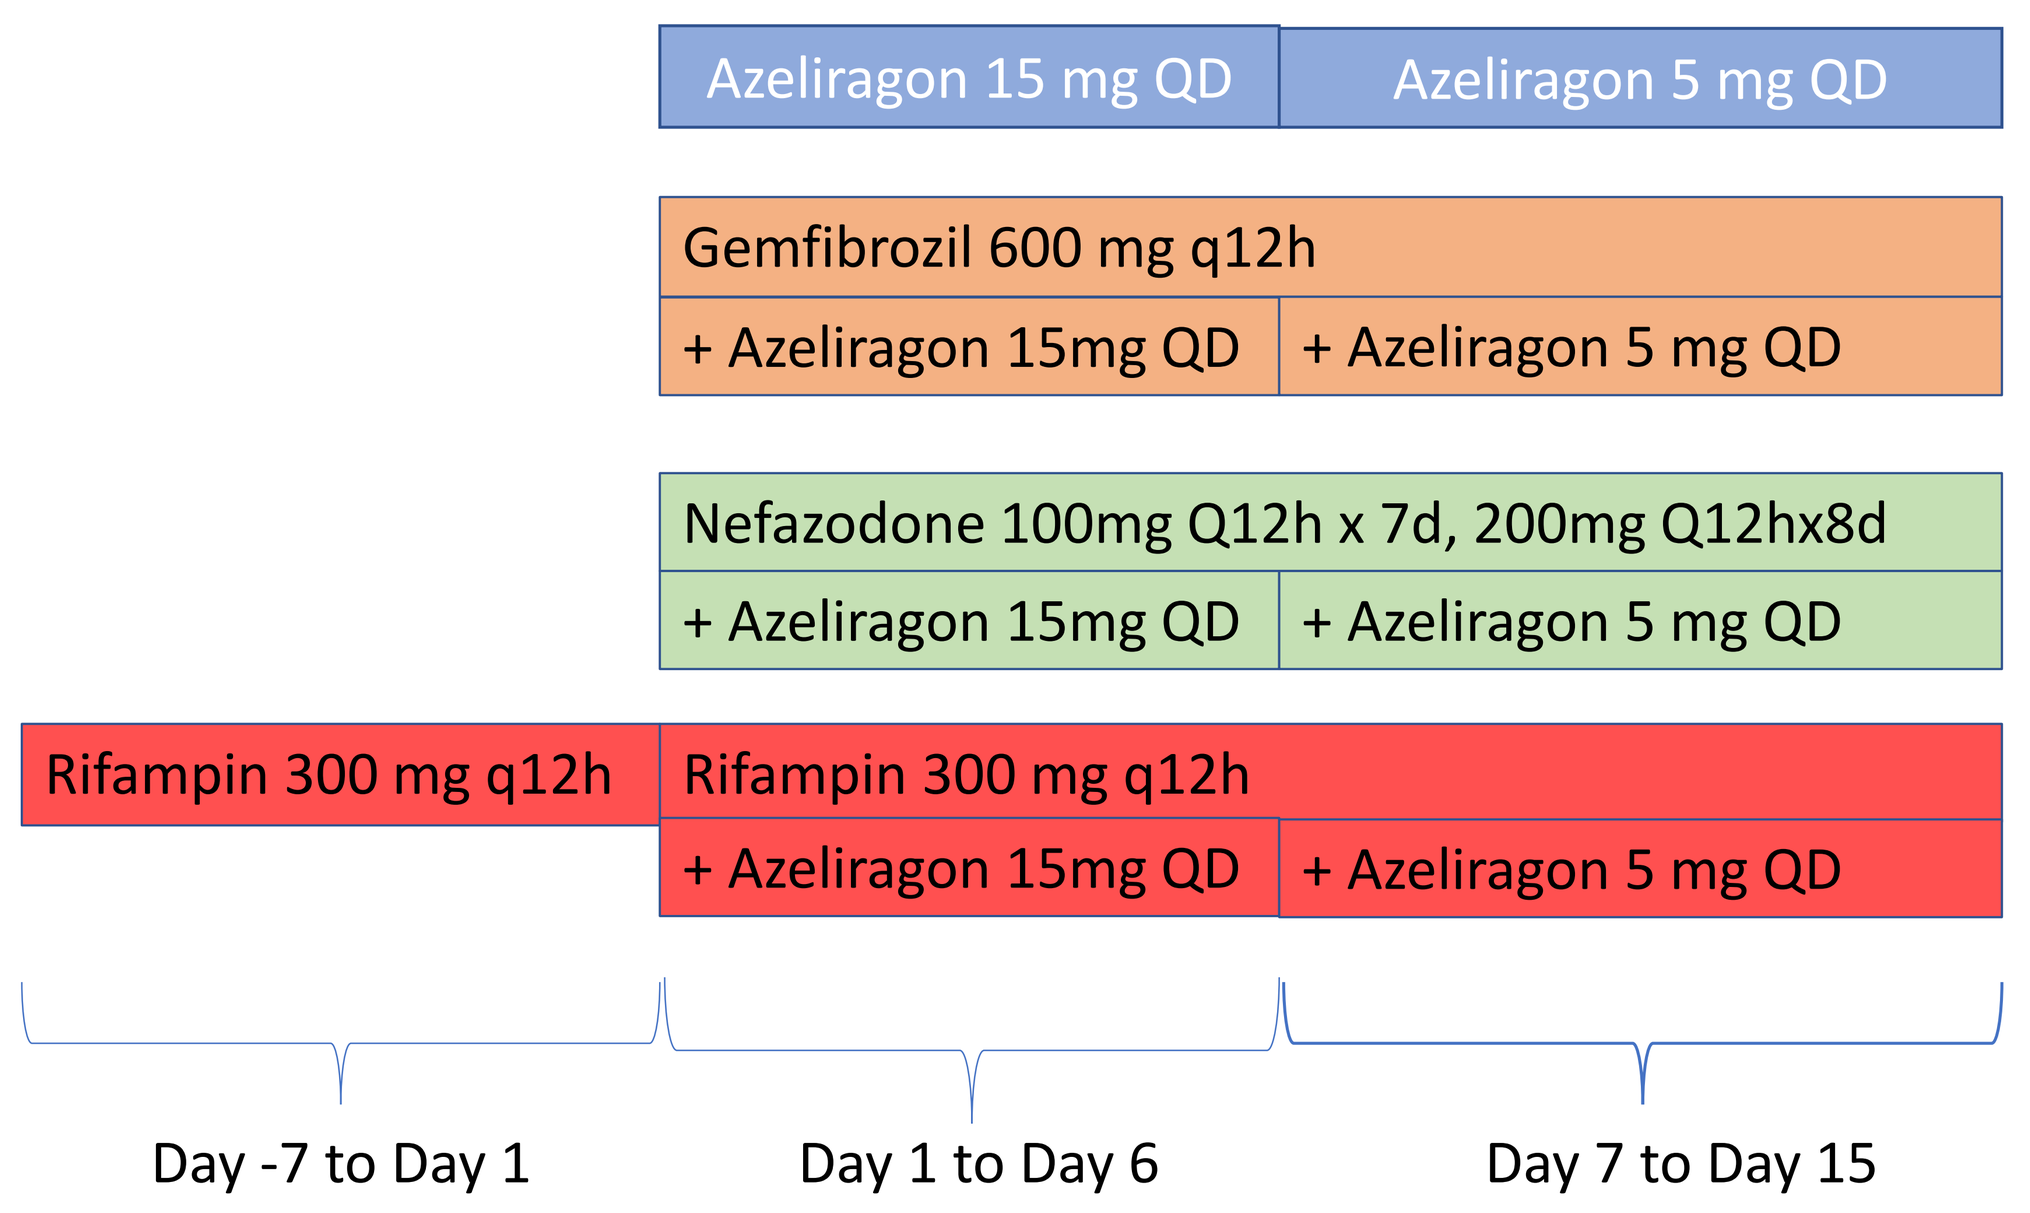
Azeliragon 15 mg QD
Azeliragon 5 mg QD
Gemfibrozil 600 mg q12h
+ Azeliragon 15mg QD
+ Azeliragon 5 mg QD
Nefazodone 100mg Q12h x 7d, 200mg Q12hx8d
+ Azeliragon 15mg QD
+ Azeliragon 5 mg QD
Rifampin 300 mg q12h
Rifampin 300 mg q12h
+ Azeliragon 15mg QD
+ Azeliragon 5 mg QD
Day -7 to Day 1
Day 1 to Day 6
Day 7 to Day 15

- Each cohort (~ 6 azeliragon alone, ~18 azeliragon + interacting medication) was admitted to the Clinical Research Unit (CRU) on Day -1 (rifampin group after 7 days pre-treatment with rifampin), underwent dosing with study medications (1h after meals) x 15d, and remained confined until Day 16.
- Blood (approximately 3 mL) for quantitating the concentrations of azeliragon and metabolites of interest in plasma were taken at:
 - Day 1: 0 (pre-dose), 0.5, 1, 2, 3, 4, 8, and 12 hours following azeliragon dosing
 - Days 2, 3, 5, 6, 7, 9, and 11: 0h (pre-dose of azeliragon)
 - Day 15: time 0 (pre-dose), 1, 4, 8, 12, 24 (Day 16), 168 (Day 22), 336 (Day 29), 504 (Day 36) and 1680 hours (Day 70) post dose.
- Plasma samples were analyzed for azeliragon, M1, M2 and M3 concentrations by validated LCMS/MS methods. The calibration range of the assays was 0.2 – 50 ng/mL. The lower limit of quantification (LLOQ) was 0.2 ng/mL.

Pharmacokinetic Analysis

Noncompartmental Analysis (NCA)

Primary Endpoints
- Cmax, Cmin, and AUCτ (where τ is 24 hours) of azeliragon on Day 15

Secondary Endpoints
- Cmax, Tmax, and AUC0-24h following the first dose of azeliragon (Day 1)
- Cmax, Cmin, and AUCτ of azeliragon metabolites of interest on Days 1 and 15
- Cmax, Cmin, and AUCτ metabolite:parent ratios for azeliragon metabolites of interest (Day 15)

Population Pharmacokinetic Analysis

Sequential modeling building process was employed using NONMEM

Primary PK endpoints
- Estimates of the popPK parameters for azeliragon (i.e., ka, Vd/F, ke, CL/F) and interaction terms, where appropriate.

Population PK Simulations
- Simulations (n=200 per treatment group) were performed to estimate the M2 and M3 plasma concentrations after dosing with azeliragon alone or after coadministration of azeliragon with gemfibrozil, rifampin, or nefazodone.
- Azeliragon and M1 appeared to have reached steady state prior to Day 15, and thus simulations were not performed for these analytes.
- PK profiles (24 hours) were simulated on Day 1, Day 15, and every 28 days from Day 28 to Day 336.
- PK parameters of the simulated profiles were estimated using a noncompartmental analysis.
- The fraction of steady state was calculated as the ratio of AUC0-24 on each day to the AUC0-24 on Day 336 for each simulation replicate.

Statistical Analysis

Noncompartmental Analysis:
- Exposure parameters (i.e., Cmax, Cmin, and AUC0-τ) were analyzed using a mixed effects statistical model, with treatment as a fixed effect and subject as a random effect.
- The geometric mean ratio and 90% confidence intervals for the ratio (test/reference) of the exposure parameters were used to estimate the effect of gemfibrozil, nefazodone, and rifampin on the PK of azeliragon and metabolites of interest.

Pop PK Analysis:
- No formal statistical analysis was performed
- Exploratory analyses include comparisons of the primary PK endpoints of azeliragon metabolites in the presence and absence of interacting drugs.

Demographics / Baseline Characteristics

Characteristic	Azeliragon (n=20)	Azeliragon + Gemfibrozil (n=18)	Azeliragon + Rifampin (n=19)	Azeliragon + Nefazodone (n=18)
Age (years)	30.1 (7.55)	29.0 (6.80)	28.6 (7.71)	33.2 (7.05)
BMI (kg/m2)	25.4 (3.73	25.6 (4.46)	25.8 (4.04)	25.1 (3.03)
Weight (kg)	81.7 (15.6)	77.9 (15.1)	77.2 (13.3)	76.9 (12.1)
Sex:				
male, n (%)	20 (100%)	17 (94.4%)	19 (100%)	17 (94.4%)
female, n (%)	0 (0%)	1 (5.56%)	0 (0%)	1 (5.56%)
Race:				
White, n (%)	8 (40.0%)	7 (38.9%)	9 (47.4%)	4 (22.2%)
Black, n (%)	11 (55.0%)	9 (50.0%)	9 (47.4%)	12 (66.7%)
Multiple, n (%)	1 (5.00%)	0 (0%)	0 (0%)	1 (5.56%)
Asian, n (%)		2 (11.1%)	1 (5.26%)	1 (5.56%)
Ethnicity:				
Not Hispanic or Latino	19 (95.0%)	16 (88.9%)	18 (94.7%)	16 (88.9%)
Hispanic or Latino	1 (5.00%)	2 (11.1%)	1 (5.26%)	2 (11.1%)

NCA Results

Mean Overlay Plot of Azeliragon Concentration vs. Time by Treatment from Day 15 to the Follow-up Visit (Semi-log Scale)

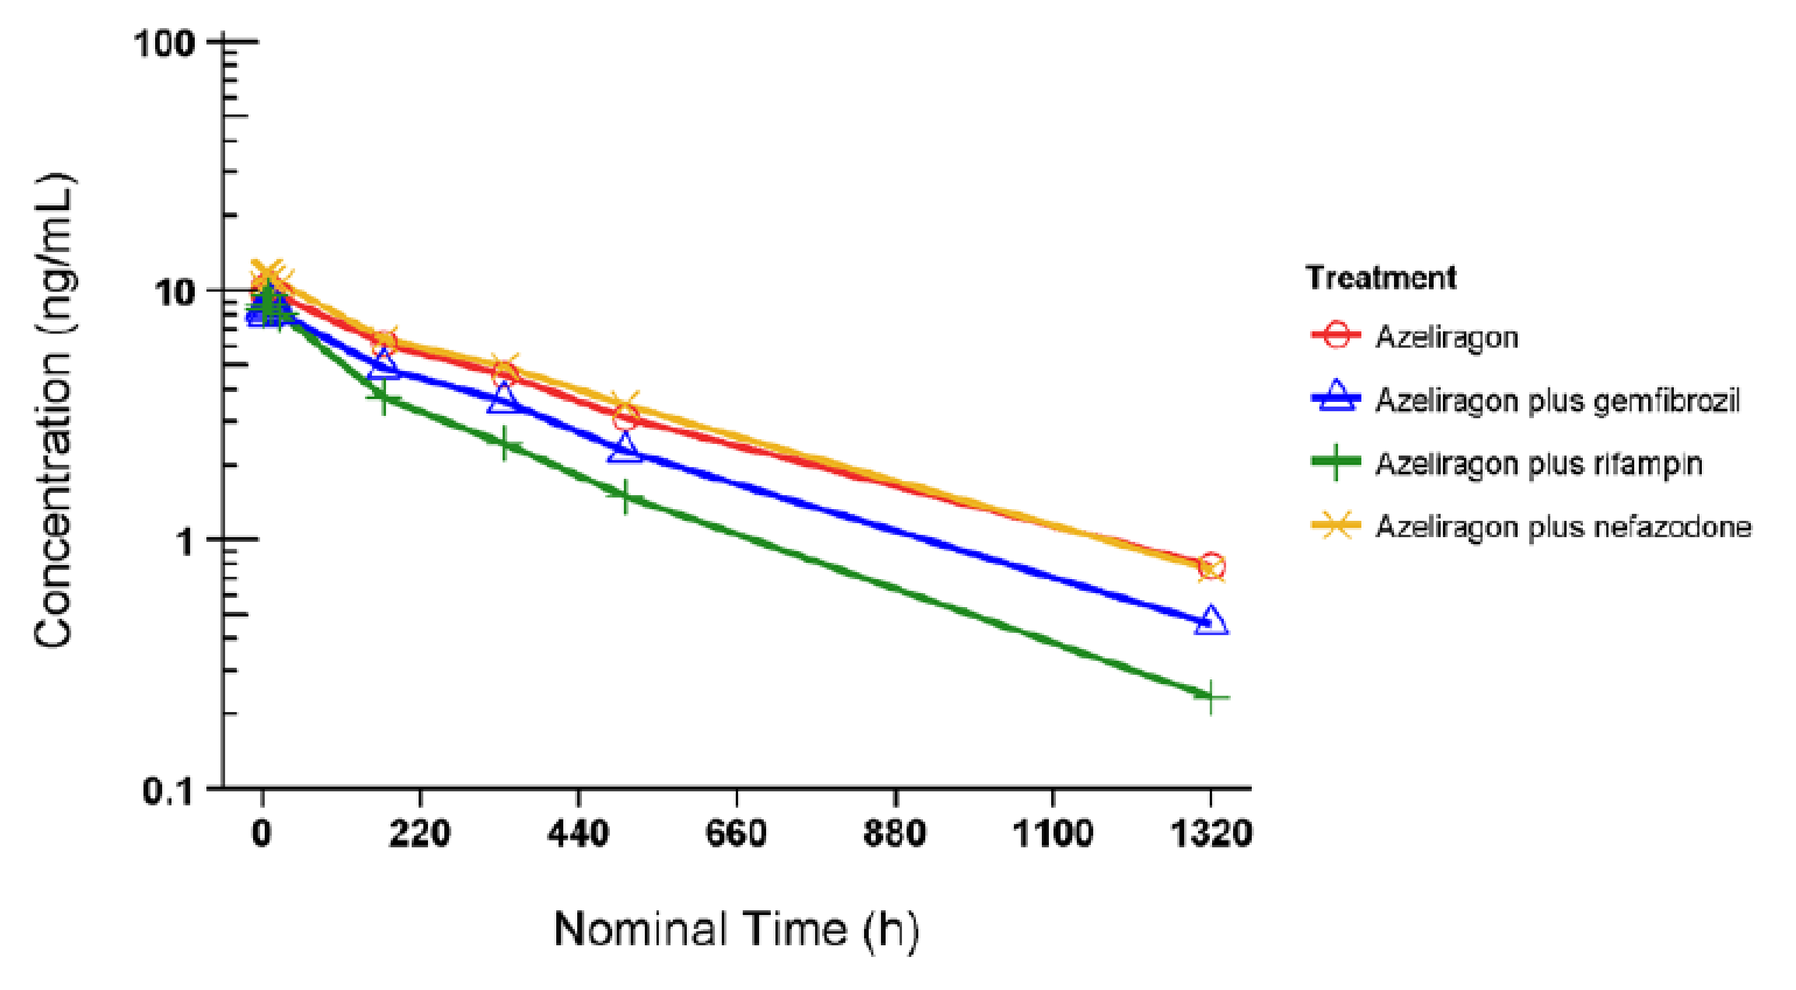
Concentration (ng/mL)
Nominal Time (h)
Treatment
Azeliragon
Azeliragon plus gemfibrozil
Azeliragon plus rifampin
Azeliragon plus nefazodone

Results of Mixed-Effects ANOVA based on Day 15 Azeliragon PK

CYP Interaction	Parameter	Geometric LSM (Test)	Geometric LSM (Reference)	Test/Reference (%)	90% Confidence Interval
CYP2C8 inhibition	Cmax (ng/mL)	8.62	10.4	82.7	(64.07, 106.75)
	AUC0-τ (h*ng/mL)	185	228	81.3	(62.53, 105.64)
	Cmin (ng/mL)	6.91	8.57	80.6	(61.82, 105.06)
CYP3A4 inhibition	Cmax (ng/mL)	11.1	10.4	106	(82.33, 137.17)
	AUC0-τ (h*ng/mL)	242	228	106	(81.79, 137.95)
	Cmin (ng/mL)	9.28	8.57	108	(83.05, 141.13)
CYP450 Induction	Cmax (ng/mL)	9.11	10.4	87.4	(67.99, 112.37)
	AUC0-τ (h*ng/mL)	195	228	85.5	(66.12, 110.61)
	Cmin (ng/mL)	7.26	8.57	84.7	(65.25, 109.97)

Mean Overlay Plot of M1 Metabolite Concentration vs. Time by Treatment from Day 15 to the Follow-up Visit (Semi-log Scale)

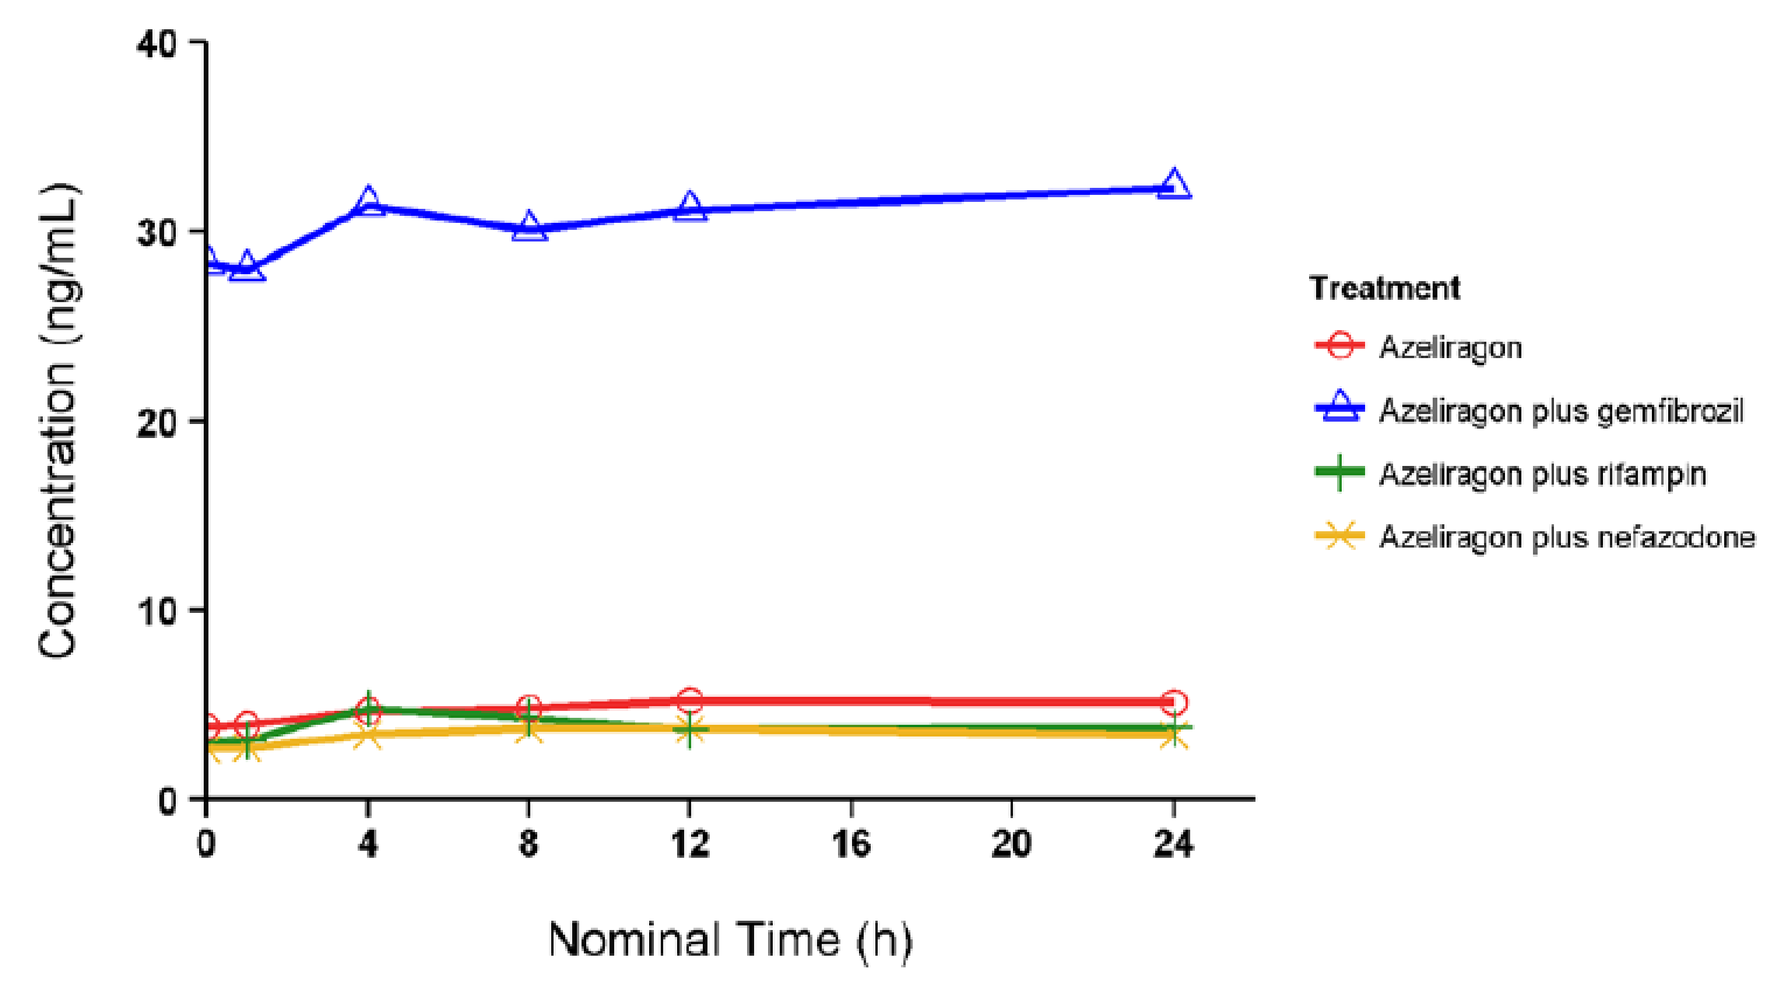
Concentration (ng/mL)
Nominal Time (h)
Treatment
Azeliragon
Azeliragon plus gemfibrozil
Azeliragon plus rifampin
Azeliragon plus nefazodone

Results of Mixed-Effects ANOVA based on Day 15 M1 metabolite PK

CYP Interaction	Parameter	Geometric LSM (Test)	Geometric LSM (Reference)	Test/Reference (%)	90% Confidence Interval
CYP2C8 inhibition	Cmax (ng/mL)	16.5	4.59	359	(214.64, 602.11)
	AUC0-τ (h*ng/mL)	356	98.9	360	(215.23, 601.80)
	Cmin (ng/mL)	12.2	3.12	392	(230.52, 667.43)
CYP3A4 inhibition	Cmax (ng/mL)	3.36	4.59	73.2	(43.69, 122.57)
	AUC0-τ (h*ng/mL)	69.2	98.9	69.9	(41.83, 116.96)
	Cmin (ng/mL)	2.14	3.12	68.5	(40.26, 116.57)
CYP450 Induction	Cmax (ng/mL)	4.22	4.59	91.9	(55.30, 152.65)
	AUC0-τ (h*ng/mL)	81.3	98.9	82.2	(49.57, 136.38)
	Cmin (ng/mL)	2.49	3.12	80.0	(47.43, 135.04)

Mean Overlay Plot of M2 Concentration vs. Time by Treatment from Day 15 to the Follow-up Visit (Semi-log Scale)

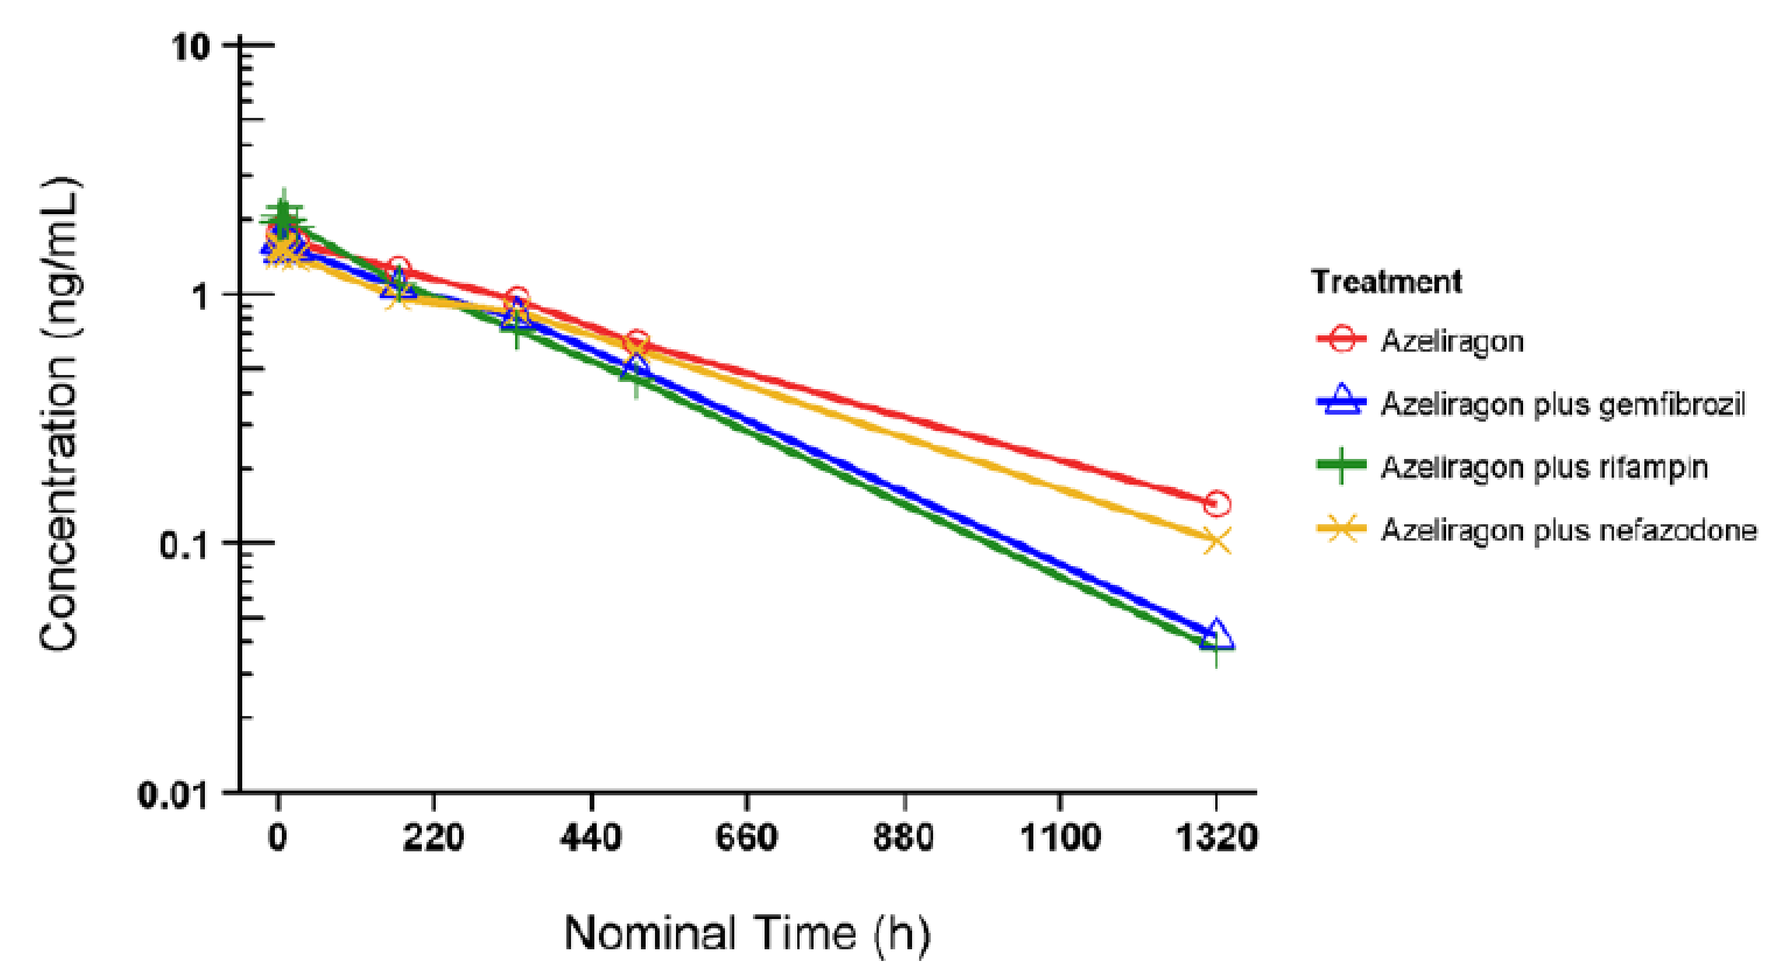
Concentration (ng/mL)
Nominal Time (h)
Treatment
Azeliragon
Azeliragon plus gemfibrozil
Azeliragon plus rifampin
Azeliragon plus nefazodone

Results of Mixed-Effects ANOVA based on Day 15 M2 metabolite PK

CYP Interaction	Parameter	Geometric LSM (Test)	Geometric LSM (Reference)	Test/Reference (%)	90% Confidence Interval
CYP2C8 inhibition	Cmax (ng/mL)	1.45	1.77	81.8	(58.50, 114.36)
	AUC0-τ (h*ng/mL)	31.1	37.9	82.1	(58.75, 114.68)
	Cmin (ng/mL)	1.16	1.41	82.4	(58.79, 115.39)
CYP3A4 inhibition	Cmax (ng/mL)	1.41	1.77	79.9	(57.13, 111.69)
	AUC0-τ (h*ng/mL)	31.1	37.9	81.9	(58.63, 114.44)
	Cmin (ng/mL)	1.19	1.41	84.4	(60.25, 118.26)
CYP450 Induction	Cmax (ng/mL)	2.06	1.77	116	(83.51, 161.56)
	AUC0-τ (h*ng/mL)	43.4	37.9	116	(82.40, 159.15)
	Cmin (ng/mL)	1.62	1.41	114	(82.00, 159.27)

Mean Overlay Plot of M3 Metabolite Concentration vs. Time by Treatment from Day 15 to the Follow-up Visit (Semi-log Scale)

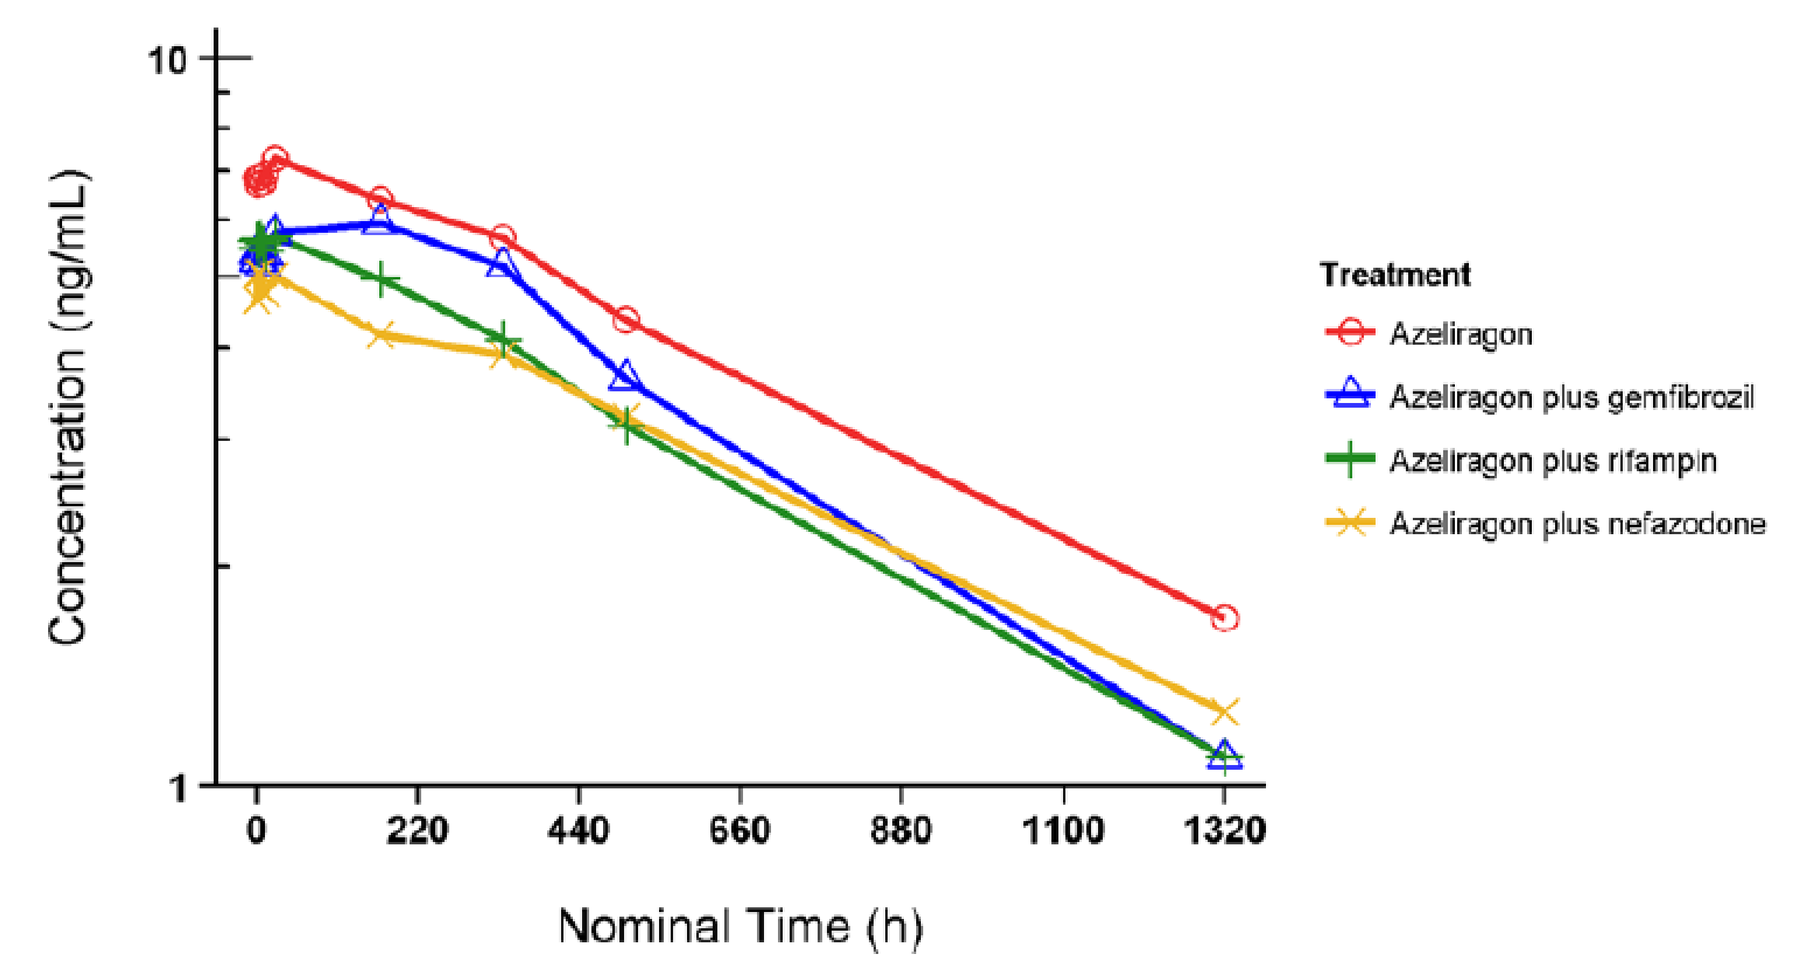
Concentration (ng/mL)
Nominal Time (h)
Treatment
Azeliragon
Azeliragon plus gemfibrozil
Azeliragon plus rifampin
Azeliragon plus nefazodone

Results of Mixed-Effects ANOVA based on Day 15 M3 metabolite PK

CYP Interaction	Parameter	Geometric LSM (Test)	Geometric LSM (Reference)	Test/Reference (%)	90% Confidence Interval
CYP2C8 inhibition	Cmax (ng/mL)	4.91	6.63	73.9	(53.32, 102.55)
	AUC0-τ (h*ng/mL)	110	149	73.7	(53.15, 102.21)
	Cmin (ng/mL)	120	149	80.1	(58.03, 110.44)
CYP3A4 inhibition	Cmax (ng/mL)	4.70	6.63	70.8	(51.09, 98.25)
	AUC0-τ (h*ng/mL)	103	149	68.9	(49.67, 95.51)
	Cmin (ng/mL)	3.94	5.74	68.6	(49.56, 95.06)
CYP450 Induction	Cmax (ng/mL)	5.30	6.63	79.8	(57.85, 110.12)
	AUC0-τ (h*ng/mL)	120	149	80.1	(58.03, 110.44)
	Cmin (ng/mL)	4.67	5.74	81.3	(58.98, 111.97)

CYP2C8 inhibition: Test = azeliragon + gemfibrozil, Reference = azeliragon alone
CYP3A4 inhibition: Test = azeliragon + nefazodone, Reference = azeliragon alone
CYP450 induction: Test = azeliragon + rifampin, Reference = azeliragon alone

Population PK Simulation Results

Conclusions of the population PK analysis are largely in line with the NCA. The largest effects seen are on M1, with gemfibrozil coadministration resulting in an 85% decrease in clearance, which is magnified by an increase in bioavailability. Predicted drug interaction on Day 15 was four-fold lower than that on Day 336, indicating that the exposure to M1 may increase nearly ten-fold when azeliragon is administered with gemfibrozil or compounds which inhibit CYP2C8 or OATP. The predicted interaction was quite variable, similar to the observed interaction on Day 15, indicating that not all patients would experience such a large increase in exposure.

Conclusions

- Azeliragon exposure was not significantly changed in the presence of CYP2C8 / 3A4 inhibitors or CYP inducer.
- Slight changes in exposure, unlikely to be clinically significant, were observed for the M2 and M3 metabolites, with the non-pharmacologically active M3 metabolite exhibiting a 4-10 fold increase.
- Data are consistent with the presence of multiple elimination pathways for azeliragon which reduces the magnitude of a clinically relevant drug-drug interaction and supports a recommendation for no requirement for azeliragon dose adjustment when co-administered with CYP3A4 inhibitors or CYP inducers.
- Co-administration with strong CYP2C8 inhibitors is not supported at this time.